Phase 1 Multiple Ascending Dose Study of CAT-2054, a Novel Oral Sterol Regulatory Element Binding Protein Inhibitor

Joanne Donovan MD PhD[1], Maria Mancini MHP[1], Andrew Nichols PhD[1]

[1]Catabasis Pharmaceuticals, Cambridge MA, USA,

Abstract

Introduction: CAT-2054, an orally administered small molecule being developed for hypercholesterolemia, is a novel inhibitor of Sterol Response Element Binding Protein (SREBP), a master regulator of lipid metabolism, including cholesterol and triglycerides, and with impact on glucose and liver fat. In CAT-2054, eicosapentaenoic acid (EPA) and nicotinic acid are conjugated by a linker that is cleaved intracellularly, producing novel pharmacology. The linker modulates the hydrolysis rate so that higher CAT-2054 levels are delivered to the liver compared to a similar compound, CAT-2003, which in clinical trials showed effects on fasting and post-prandial triglycerides as well as LDL-C and glucose. In preclinical studies CAT-2054 undergoes efficient cellular uptake and delivery to the endoplasmic reticulum, where inhibition of SREBP activation prevents induction of key genes involved in cholesterol metabolism and ultimately reduces LDL-C. In nonhuman primates, CAT-2054 reduced LDL-C over a 6 week dosing period.

Purpose: To assess safety, pharmacokinetics (PK) and pharmacodynamics of CAT-2054 in a multiple ascending dose study.

Methods: Doses of 100-750 mg QD, and 250 mg BID of CAT-2054 or matching placebo were administered for 14 days to normal healthy volunteers (N=70) aged 18-55 with LDL-C≥100 mg/dL. CAT-2054 and its metabolites in plasma was measured over 24 hours. Additionally CAT-2054 500 mg QD with 40 mg atorvastatin was given to assess PK in combination with CAT-2054 over 14 days. Safety was evaluated by adverse events, laboratories (chemistry, hematology, coagulation), physical examination and EKG. Plasma lipids were assessed after 14 days of treatment.

Results: CAT-2054 was well-tolerated and there were no serious adverse events or discontinuations. Following CAT-2054 administration, no safety signals in laboratories, vital signs or electrocardiogram results were observed, and there was no evidence of flushing. At the highest doses, the most common adverse events were gastrointestinal. PK showed dose-proportional increases in plasma CAT-2054 concentrations and there was no evidence for impact on atorvastatin PK. At Day 14, median decreases in LDL-C of up to 11% were observed. At Day 21, there were decreases in LDL-C of up to 20% in all dosing groups.

Conclusions: In this initial clinical study, CAT-2054 lowered LDL-C and was well tolerated with no safety signals. Consistent with the intent to modulate the hydrolysis rate to deliver higher levels to the liver, CAT-2054 had greater systemic exposure than CAT-2003. CAT-2054 is being studied in a Phase 2 trial to inhibit SREBP, the master regulator of lipid metabolism, for patients with hypercholesterolemia.

Disclosure of conflicts of interest:
Joanne M. Donovan, MD PhD, Chief Medical Officer
Maria Mancini, MHP, Senior Director, Clinical Operations
Andrew Nichols, PhD, SVP of Research and Non-Clinical Development

All are employees of and hold stock and stock options in Catabasis Pharmaceuticals, Inc.

Background

Background

- Sterol Regulatory Element Binding Protein (SREBP) is a master regulator of lipid metabolism and controls the metabolism of both LDL-C and triglycerides, through effects on multiple downstream proteins including PCSK9 and HMGCoA reductase, making it a potential therapeutic target for nonalcoholic steatohepatitis (NASH) and for hypercholesterolemia.
- CAT-2000 compounds are novel inhibitors of the SREBP transcription factor system. In this series of compounds eicosapentaenoic acid (EPA) and nicotinic acid are conjugated by a linker that is cleaved by the intracellular enzyme fatty acid amide hydrolase (FAAH).
 - First generation compound, CAT-2003, targeting intestine, reduces triglycerides, LDL-C and HbA1c (Donovan, 2015, NLA)
- CAT-2054 is a novel oral second-generation product candidate targeted to the liver in development for NASH and hypercholesterolemia.

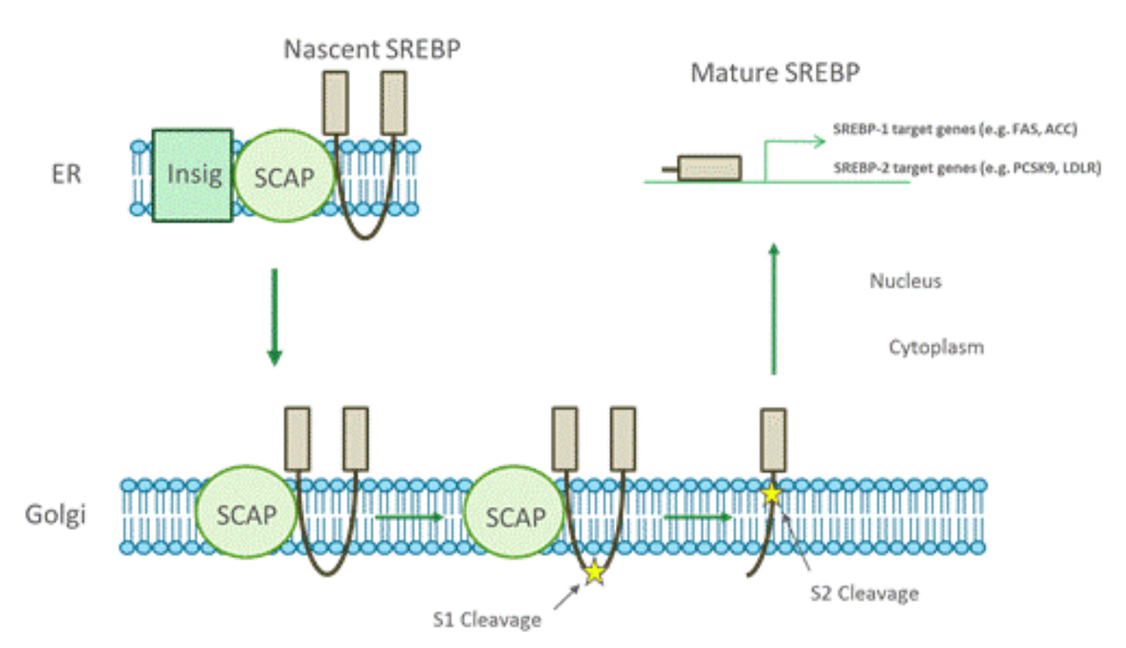
Nascent SREBP
Mature SREBP
ER
Insig
SCAP
Nucleus
Cytoplasm
Golgi
S1 Cleavage
S2 Cleavage

CAT-2003 and CAT-2054 are First in Class Inhibitors of SREBP, a Master Regulator of Lipid Metabolism

First in class SREBP inhibitors, acting on SREBP1 to inhibit key de novo lipogenesis genes, and acting on SREBP2 to inhibit key cholesterol synthesis genes

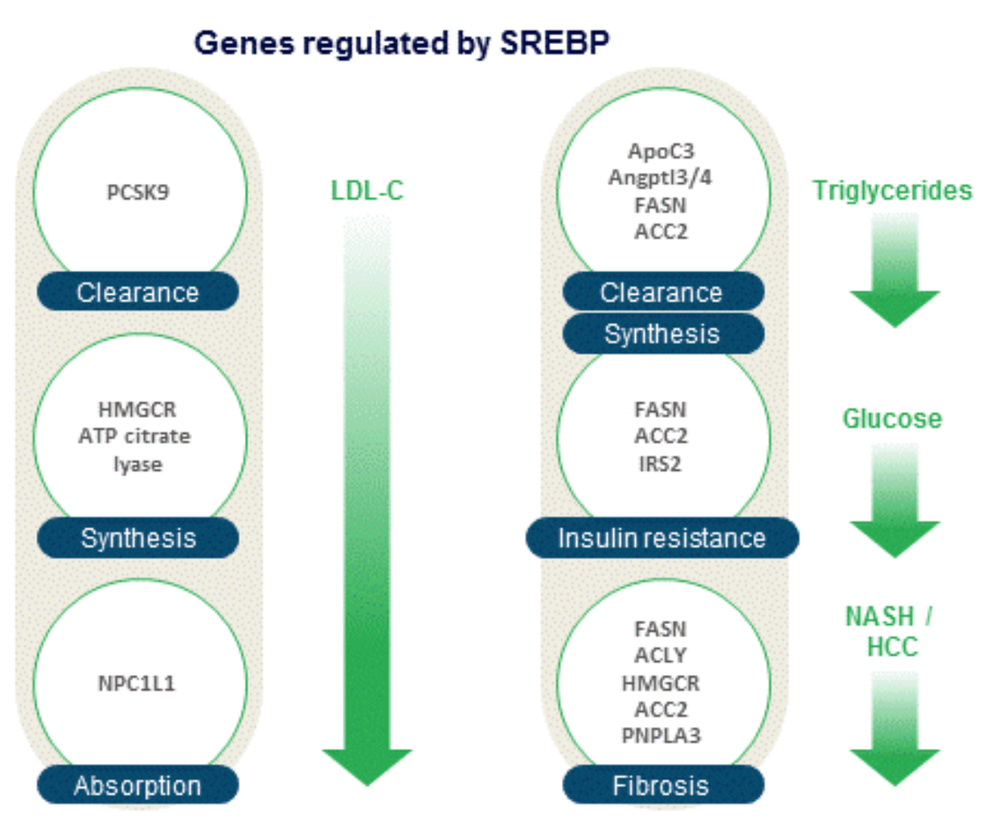
Genes regulated by SREBP
PCSK9
Clearance
HMGCR
ATP citrate lyase
Synthesis
NPC1L1
Absorption
LDL-C
ApoC3
Angptl3/4
FASN
ACC2
Clearance
Synthesis
FASN
ACC2
IRS2
Insulin resistance
FASN
ACLY
HMGCR
ACC2
PNPLA3
Fibrosis
Triglycerides
Glucose
NASH / HCC

CAT-2003 & CAT-2054 Inhibit SREBP-1 and SREBP-2 Targets

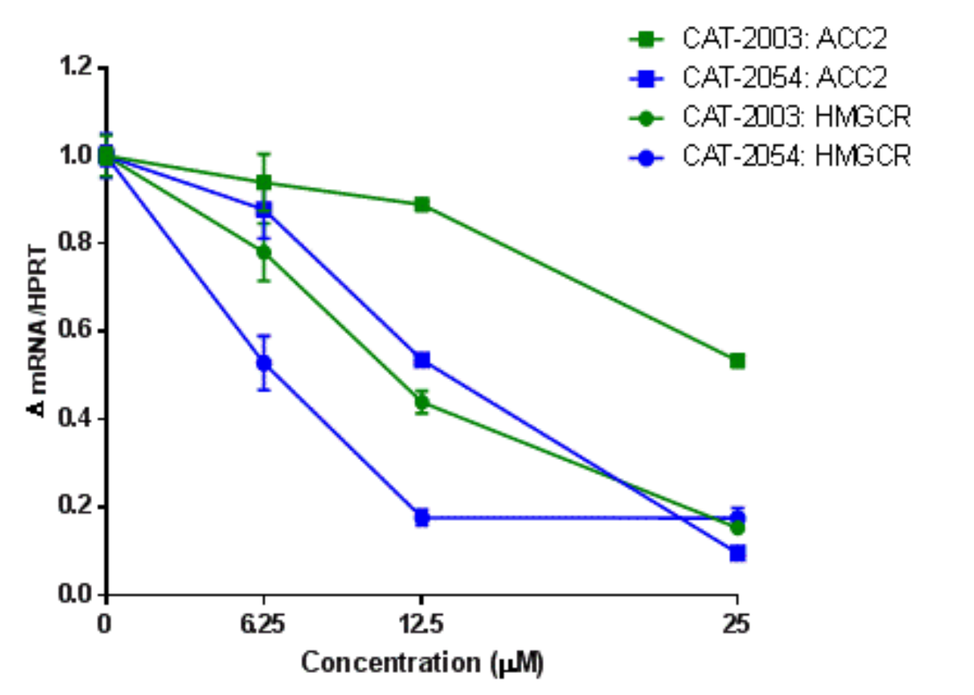
CAT-2003: ACC2
CAT-2054: ACC2
CAT-2003: HMGCR
CAT-2054: HMGCR
Δ mRNA/HPRT
Concentration (μM)

Picarella et al, 2016 DDW
HepG2 cells treated with indicated concentrations of CAT-2003 or CAT-2054. Cells were harvested the next day for mRNA analysis. Error bars represent SEM.

Proof of Concept for SREBP Modulation with First-Generation Conjugate CAT-2003 Phase 2 Data

CAT-2003 Reductions in Fasting Triglycerides, LDL-C and Glucose at Week 4

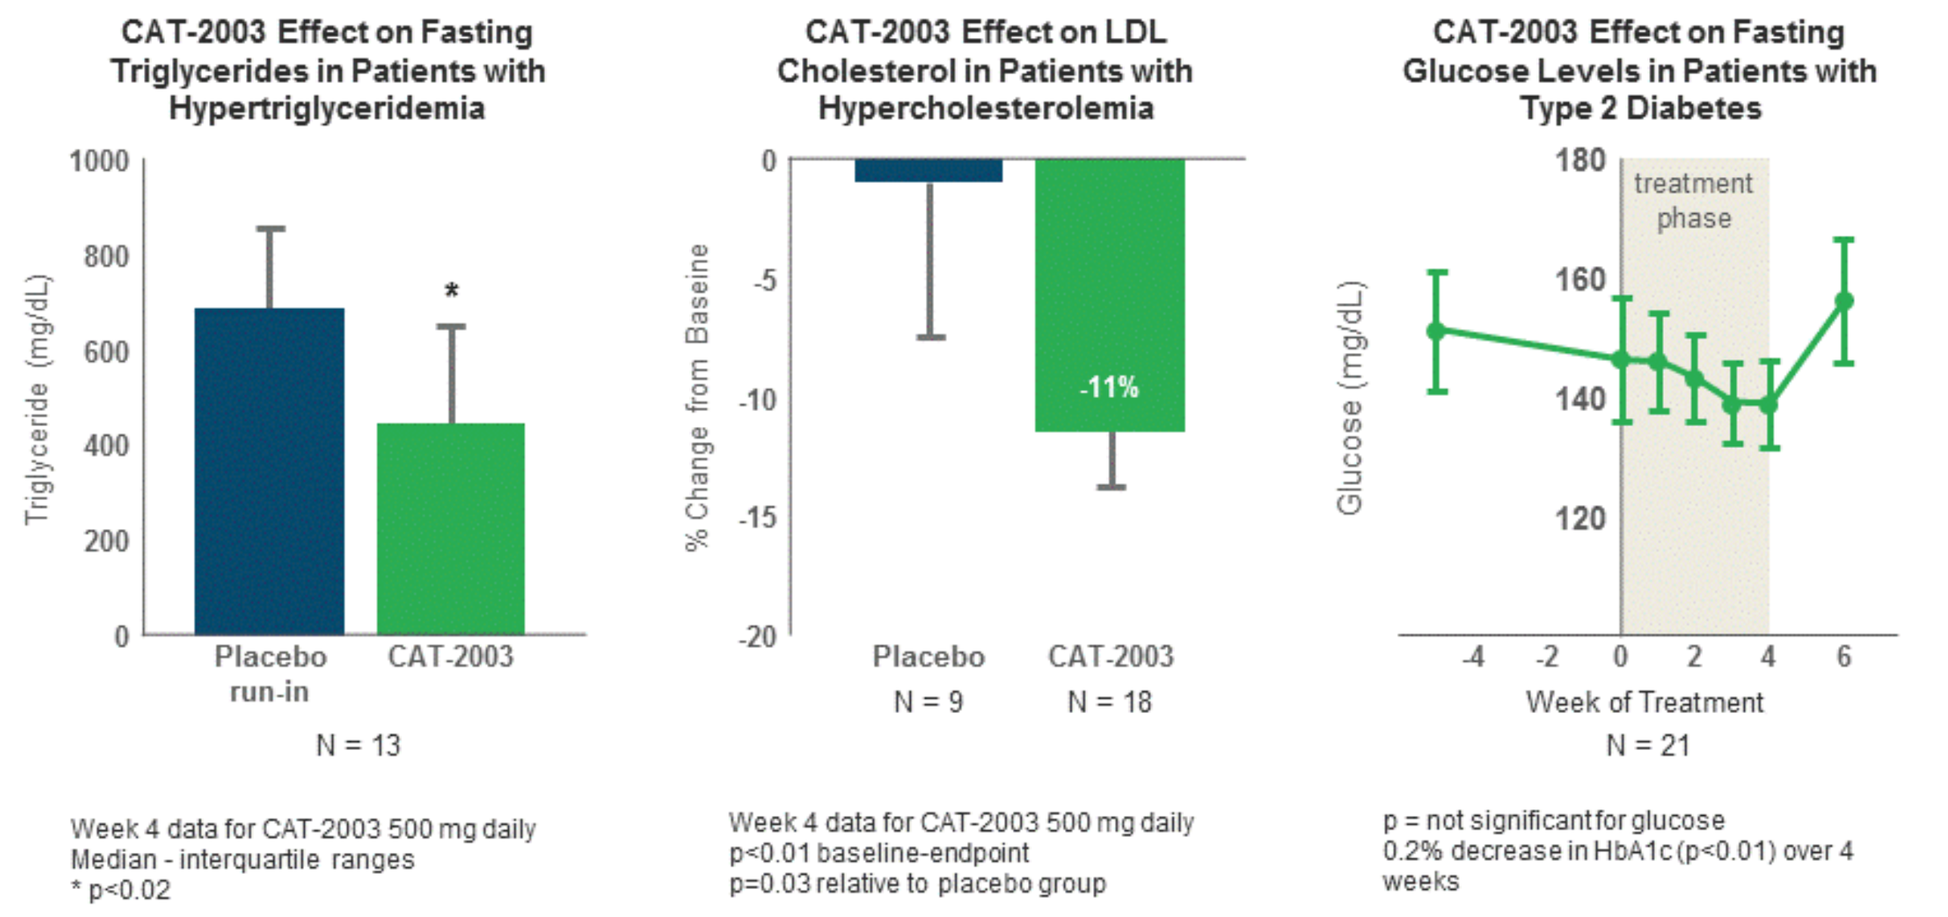
CAT-2003 Effect on Fasting Triglycerides in Patients with Hypertriglyceridemia
Triglyceride (mg/dL)
Placebo run-in
CAT-2003
N = 13
CAT-2003 Effect on LDL Cholesterol in Patients with Hypercholesterolemia
% Change from Baseine
-11%
Placebo
N = 9
CAT-2003
N = 18
CAT-2003 Effect on Fasting Glucose Levels in Patients with Type 2 Diabetes
treatment phase
Glucose (mg/dL)
Week of Treatment
N = 21

Week 4 data for CAT-2003 500 mg daily
Median - interquartile ranges
* p<0.02

Week 4 data for CAT-2003 500 mg daily
p<0.01 baseline-endpoint
p=0.03 relative to placebo group

p = not significant for glucose
0.2% decrease in HbA1c (p<0.01) over 4 weeks

Phase 1 Single Ascending Dose Study of CAT-2054

- CAT-2054 was designed to be stable to intracellular enzymatic cleavage to enable more intact CAT-2054 to pass through the portal vein and to the liver, where SREBP controls cholesterol levels and modulates hepatic lipid synthesis.
- In the phase 1 single ascending dose study of doses from 50 to 1000 mg, CAT-2054 was well-tolerated
 - No safety signals in laboratory, vital signs or electrocardiogram results
 - Most common AEs observed in fed and fasting conditions were mild nausea and diarrhea at highest dose.

CAT-2054 Plasma Exposure Exceeds that for CAT-2003

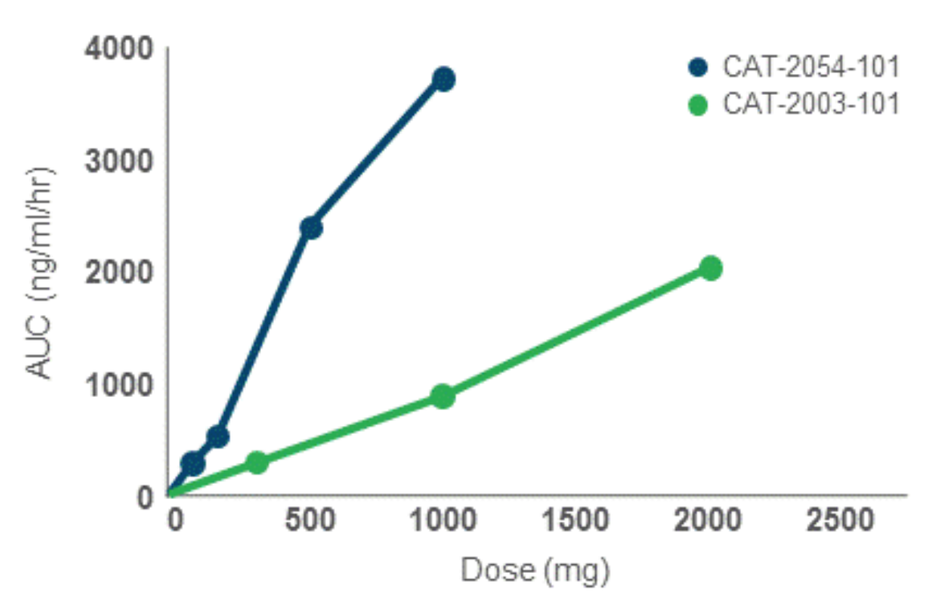
CAT-2054-101
CAT-2003-101
AUC (ng/ml/hr)
Dose (mg)

CAT-2054 and CAT-2003 data from independent Single Ascending Dose trials

Inhibition of SREBP with CAT-2054 Analog (CAT-2003) Reduces Steatohepatitis in ApoE*3 Leiden mice

Liver Histopathology

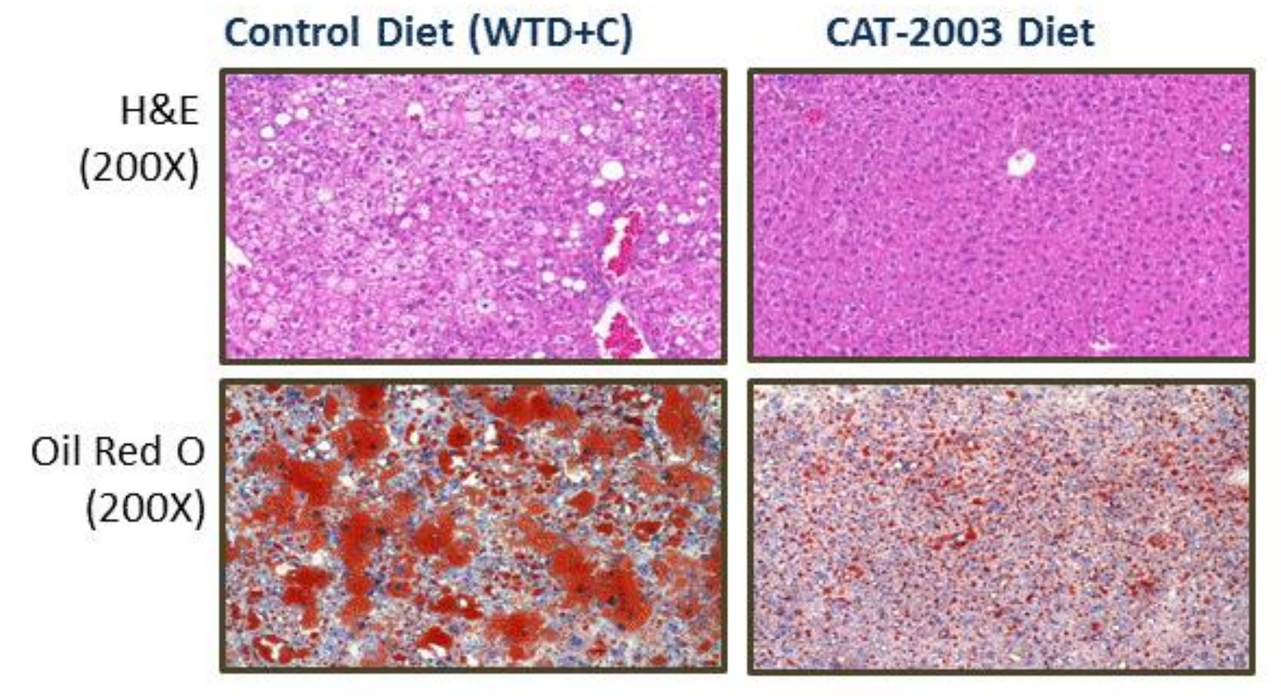
Control Diet (WTD+C)
CAT-2003 Diet
H&E (200X)
Oil Red O (200X)

16 wk, apoE*3 Leiden mice. 0.75% CAT-2003 diet admix (~125 mg/kg equivalent dose). Error bars represent SEM (n=16).

Zimmer et al., ATVB, 2015

Inhibition of SREBP with CAT-2054 Analog (CAT-2003) Decreases Liver Fat, Inflammation and Fibrosis in NASH Model

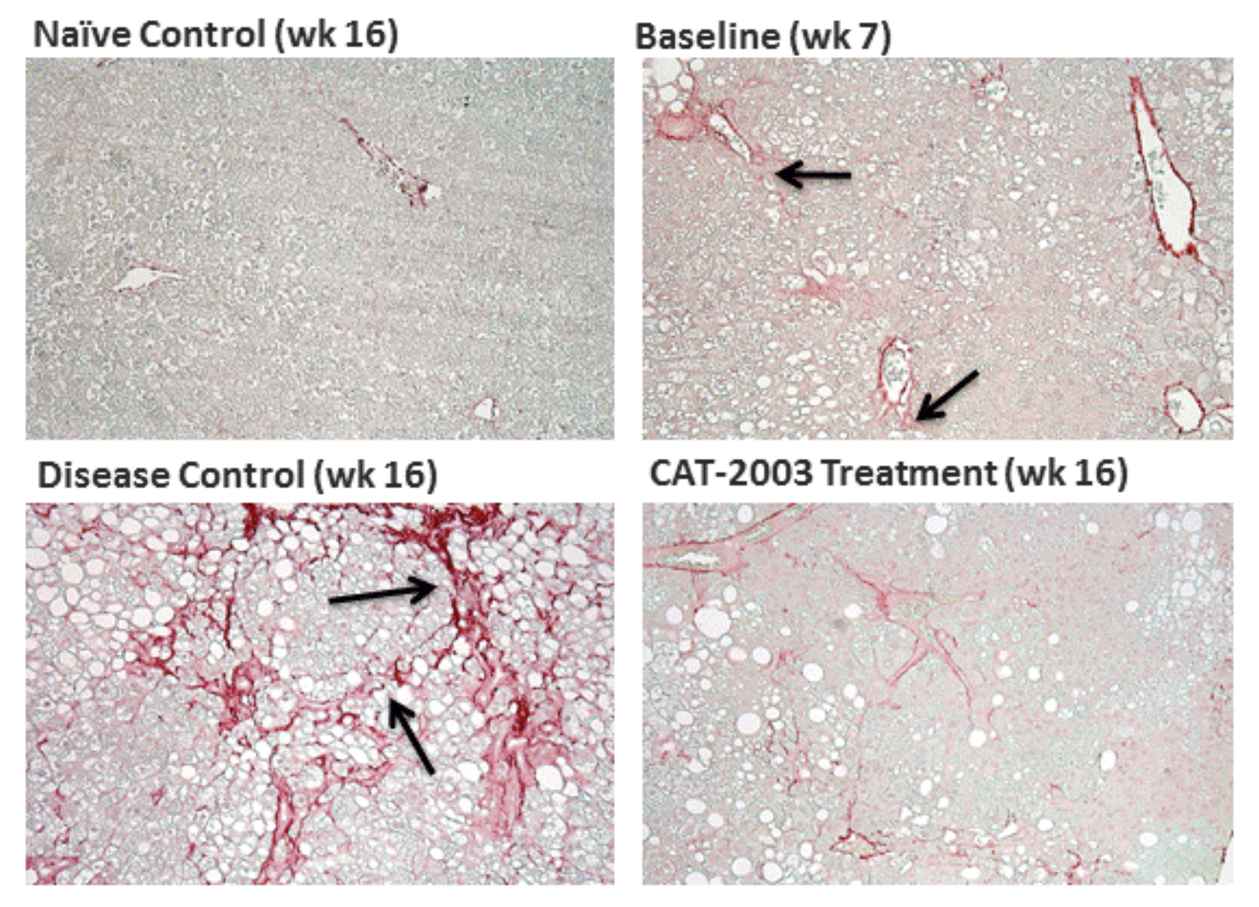
Naïve Control (wk 16)
Baseline (wk 7)
Disease Control (wk 16)
CAT-2003 Treatment (wk 16)

A two hit model was used to induce a NASH-like disease in male C57BL/6 mice. Streptozotocin was used to induce liver and pancreatic damage in 4 day old mice followed by a second insult of a high fat/cholesterol diet at week 5 to induce steatohepatitis and fibrosis. CAT-2003 was administered in the diet (0.75% w/w) 2 weeks later for 9 weeks. CAT-2003 reduced the fibrosis score from 1.83 in the disease control to 1.17 which was lower than the pretreatment baseline score of 1.38.

- In a metabolic murine model of NASH, control mice developed characteristic features of NASH including hepatic steatosis, lobular inflammation, ballooning degeneration, and fibrosis
- CAT-2003 treatment resulted in decreased expression of genes involved in fibrosis,
- CAT-2003 treatment decreased the NAFLD Activity Score (NAS), hepatic cholesterol levels, fibrosis and overall severity of pre-neoplastic

Picarella et al., DDW, 2016

Results: Phase 1

Objectives and Study Design

- **Objective**
 - To assess the safety, tolerability, pharmacokinetics and pharmacodynamics of CAT-2054 administered for 14 days over a range of doses
 - To support design of Phase 2 study by assessing pharmacokinetics of atorvastatin when coadministered with CAT-2054 for 14 days
- **Study design**
 - Multiple ascending doses
 - Randomized, double-blind, placebo-controlled (N=8-10 per cohort)
 - Doses: 100, 250, 400, 500, 750 mg QD and 250 mg BID PO
 - Atorvastatin PK
 - Single dose of atorvastatin 40 mg, to be compared with CAT-2054 500 mg and atorvastatin 40 mg coadministered for 14 days (N =10), powered to demonstrate that confidence intervals of AUC of atorvastatin coadministered with CAT-2054 were between 50 and 200%
- **Population:**
 - Normal healthy volunteers aged 18-55 with LDL-C≥100 mg/dL
- **Study Drug:**
 - CAT-2054 or placebo in soft-gel capsules
- **Key Endpoints**
 - Safety and tolerability
 - Pharmacokinetics
 - Pharmacodynamics

CAT-2054: Effects on LDL-C

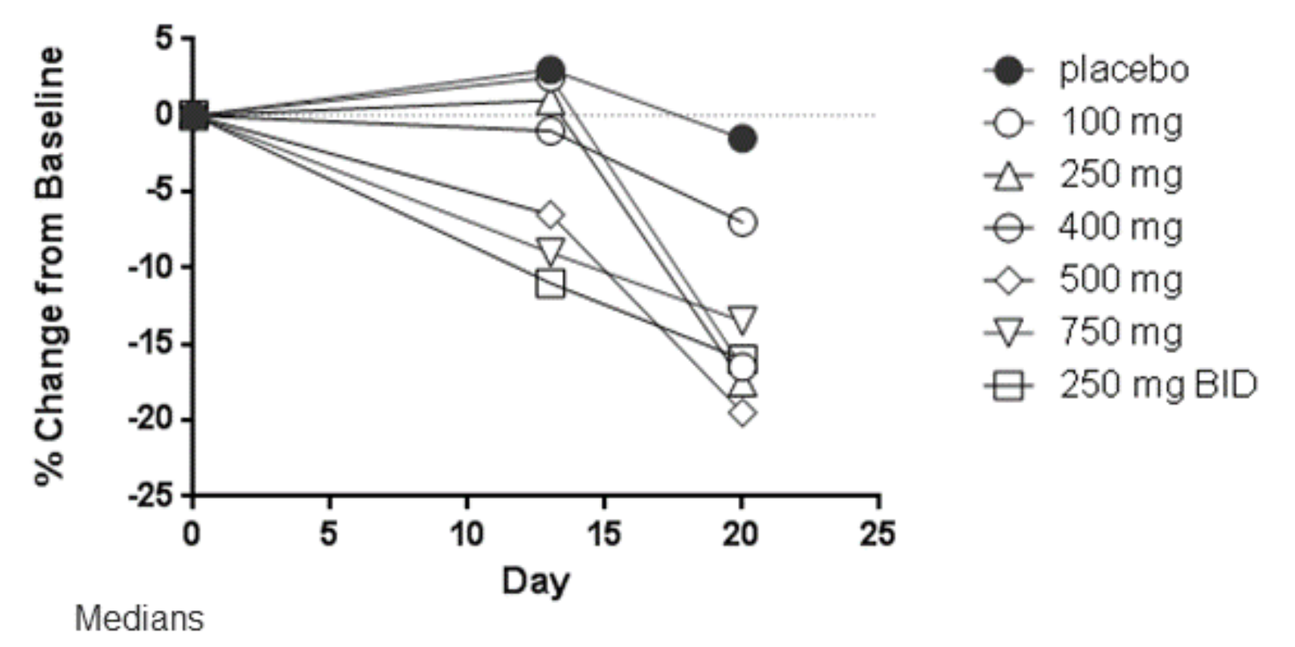
% Change from Baseline
Day
placebo
100 mg
250 mg
400 mg
500 mg
750 mg
250 mg BID

Medians

- CAT-2054 changes except BID from baseline to 21 days p<0.05, placebo NS
- Endpoint mean of Day 14-15 and Baseline Day 1

- Dose responsive LDL-C reduction at end of dosing at Day 14
- Further LDL-C reductions after Day 14 that were statistically significant at Day 21 for multiple dose groups
- Changes in PCSK9 were not observed

CAT-2054 Safety Overview

- No trends in safety labs, ECG, physical exams
- Adverse events in >1 subject, mild (except 1 mod), no discontinuations

Preferred term	Placebo (n=12)	CAT-2054						
		100 mg (n=8)	250 mg (n=8)	400 mg (n=8)	250 mg BID(n=8)	500 mg (n=8)	750 mg (n=8)	500 mg /Ator (n=10)
Diarrhoea	1	0	0	2	1	4	4	3
Abdominal pain	0	0	1	0	0	1	2	4
Headache	3	0	0	0	1	1	1	0
Nausea	0	0	0	0	1	0	1	3
Dyspepsia	0	0	0	1	0	0	1	3
Hyperhidrosis	0	0	0	0	0	1	2	0
Constipation	0	0	0	0	0	2	0	0
Somnolence	1	0	0	0	0	0	1	0
Vomiting	0	0	0	0	0	0	1	1
Erythema	0	0	0	0	0	2	0	0

Pharmacokinetics of Coadministered CAT-2054 and Atorvastatin

- Pharmacokinetics of CAT-2054 coadmininistered with atorvastatin 40 mg for 14 days
 - Powered to show that 95% confidence interval was between 50 and 200%, i.e., no strong Cyp3A4 inhibition to increase atorvastatin 40 mg levels above 80 mg level
 - Accumulation ratio of atorvastatin 40 mg ~1.3 (Cilla, 1995, Clinical Pharm Therapeutics)
 - No clinically relevant effect of atorvastatin 40 mg on Day 14 PK of CAT-2054

Ratio of AUC_{0-48} on Day 14/ Day -2: Mean and 95% CI

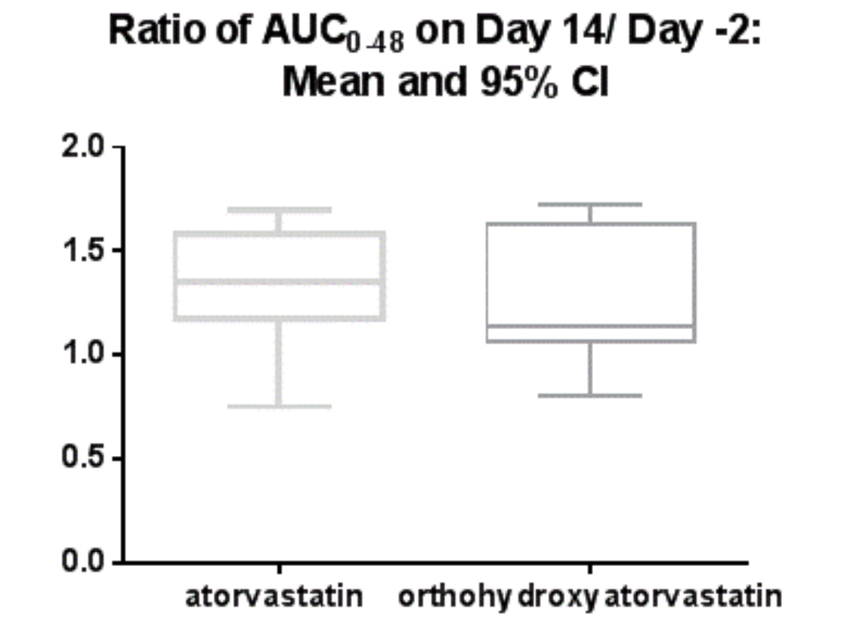
atorvastatin
orthohydroxy atorvastatin

Conclusion:
- *No evidence for clinically significant impact of CAT-2054 on atorvastatin PK*

Multiple Dose Pharmacokinetics of CAT-2054

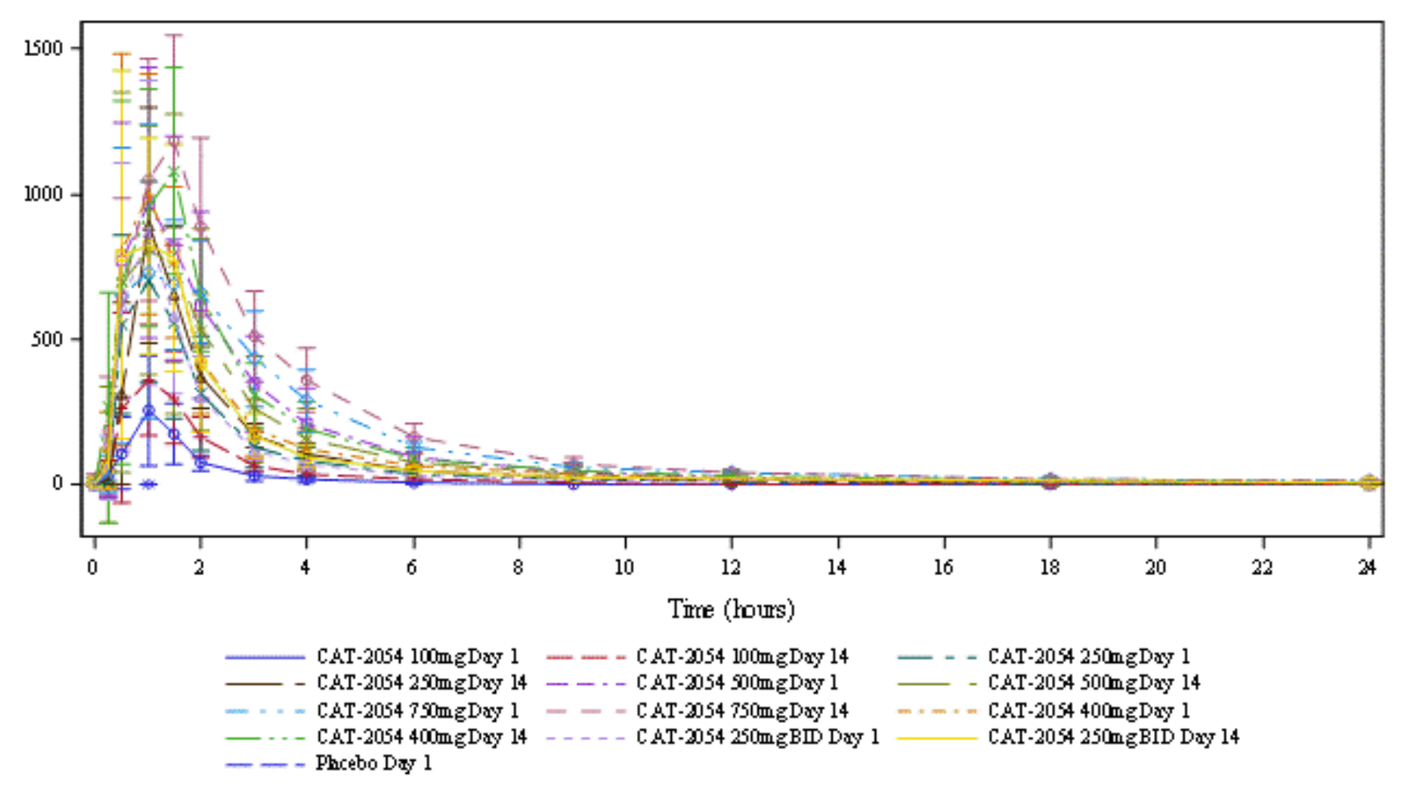
Concentration (ng/mL)
Time (hours)
CAT-2054 100mg Day 1
CAT-2054 100mg Day 14
CAT-2054 250mg Day 1
CAT-2054 250mg Day 14
CAT-2054 500mg Day 1
CAT-2054 500mg Day 14
CAT-2054 750mg Day 1
CAT-2054 750mg Day 14
CAT-2054 400mg Day 1
CAT-2054 400mg Day 14
CAT-2054 250mg BID Day 1
CAT-2054 250mg BID Day 14
Placebo Day 1

- Pharmacokinetics of CAT-2054:
 - Approximately dose proportional
 - Minimal accumulation over 14 days
 - Metabolites EPA and nicotinic acid not observed over background levels
 - Major detected metabolite nicotinuric acid

Conclusions and Future Directions

- In this initial 14-day multiple dose clinical study CAT-2054 was well tolerated with no safety signals.
- CAT-2054 lowered LDL-C after 14 days of dosing QD or BID, with greatest reductions at Day 21
- Coadministration of CAT-2054 and atorvastatin 40 mg did not result in clinically relevant effects on either CAT-2054 or atorvastatin pharmacokinetics
- CAT-2054 is a novel oral product candidate in development for NASH and hypercholesterolemia, based on targeting SREBP. CAT-2054 is currently in a Phase 2 trial in patients with hypercholesterolemia who are on atorvastatin 40 mg daily.
- Targeting SREBP has the potential to decrease LDL-C and have positive effects on NASH, a disease associated with increased cardiometabolic risk

catabasis
a return to health