CAT-2003, an Analog of CAT-2054, a Novel Oral Sterol Regulatory Element Binding Protein Inhibitor, Inhibits Inflammation and Fibrosis in a Murine Model of Nonalcoholic Steatohepatitis (NASH)

Dominic Picarella[1], Mike Zimmer[1], Diana Lee[1], Michael Briggs[2], Arlin Rogers[3], Joanne Donovan[1] and Andrew Nichols[1]

[1]Catabasis Pharmaceuticals, Inc., [2]Woodland Biosciences, [3]Cummings School of Veterinary Medicine Tufts University

catabasis

Abstract

Introduction: CAT-2003 and CAT-2054 are orally administered small molecule inhibitors of Sterol Response Element-Binding Protein (SREBP), a master regulator of cholesterol and triglyceride metabolism that impacts liver fat and cholesterol levels. Consistent with this mechanism CAT-2003 and CAT-2054 treatment reduced LDL cholesterol levels in clinical trials. Elevated expression of SREBP isoforms have been described in patients with NAFLD and NASH. CAT-2054 is currently in a Phase 2a trial for hypercholesterolemia. Here, we use CAT-2003 as an analog molecule of CAT-2054 to study effects in a murine model of NASH.

Objective: To determine the effect of oral administration of CAT-2003 on the progression of inflammation, fibrosis and NASH and the subsequent development of pre-neoplastic lesions in a murine model.

Methods: A two hit model was used to induce a NASH-like disease in male C57BL/6 mice. Streptozotocin was used to induce liver and pancreatic damage in 4 day old mice followed by a second insult of a high fat/cholesterol diet at week 5 to induce steatohepatitis and fibrosis. CAT-2003 was administered in the diet (0.75% w/w) 2 weeks later for 9 weeks.

Results: CAT-2003 treatment significantly reduced hepatic INSIG1 expression, indicating inhibition of SREBP activity. CAT-2003 significantly reduced steatosis and inflammation and produced a complete abrogation of the ballooning degeneration and progression of fibrosis relative to levels observed in baseline control mice at the start of treatment. The absolute NAFLD/NASH activity score was significantly reduced from 4.75 in control to 2.83 in treated animals. Similarly, CAT-2003 reduced the fibrosis score from 1.83 in the disease control to 1.17 which was lower than the pretreatment baseline score of 1.38. Livers from CAT-2003 treated animals showed markedly decreased expression of the pro-inflammatory and pro-fibrogenic chemokines CCL2 and CCL20; ACTA2, a marker of hepatic stellate cell activation; and ID1, an indicator of reduced TGFβ signaling. In addition, CAT-2003 also significantly reduced the number of pre-neoplastic foci from a mean of 6.8 to 2.0/cm^2, demonstrating that treatment with CAT-2003 may help slow progression to hepatocellular carcinoma.

Conclusions: Inhibition of SREBP by oral administration of CAT-2003, an analog of CAT-2054, significantly reduced the progression of multiple morphologic components of NASH including: steatosis, inflammation, hepatocyte injury and fibrosis. CAT-2054, which is in a Phase 2a trial for hypercholesterolemia, may therefore have additional utility in NASH.

Disclosure of conflicts of interest:
Dominic Picarella, PhD, Director of Pharmacology, Mike Zimmer PhD, Principal Scientist, Diana Lee, Associate Scientist, Joanne M. Donovan, MD PhD, Chief Medical Officer, Andrew Nichols, PhD, SVP of Research and Non-Clinical Development

All are employees of and hold stock and stock options in Catabasis Pharmaceuticals, Inc.

Background

Pathway for SREBP Maturation

- Sterol Regulatory Element Binding Protein (SREBP) is a master regulator of lipid metabolism and controls the metabolism of both LDL-C and triglycerides, through effects on multiple downstream proteins including PCSK9 and HMGCoA reductase.
- CAT-2000 compounds are novel inhibitors of the SREBP transcription factor system. In this series of compounds eicosapentaenoic acid (EPA) and nicotinic acid are conjugated by a linker that is cleaved by the intracellular enzyme fatty acid amide hydrolase (FAAH).

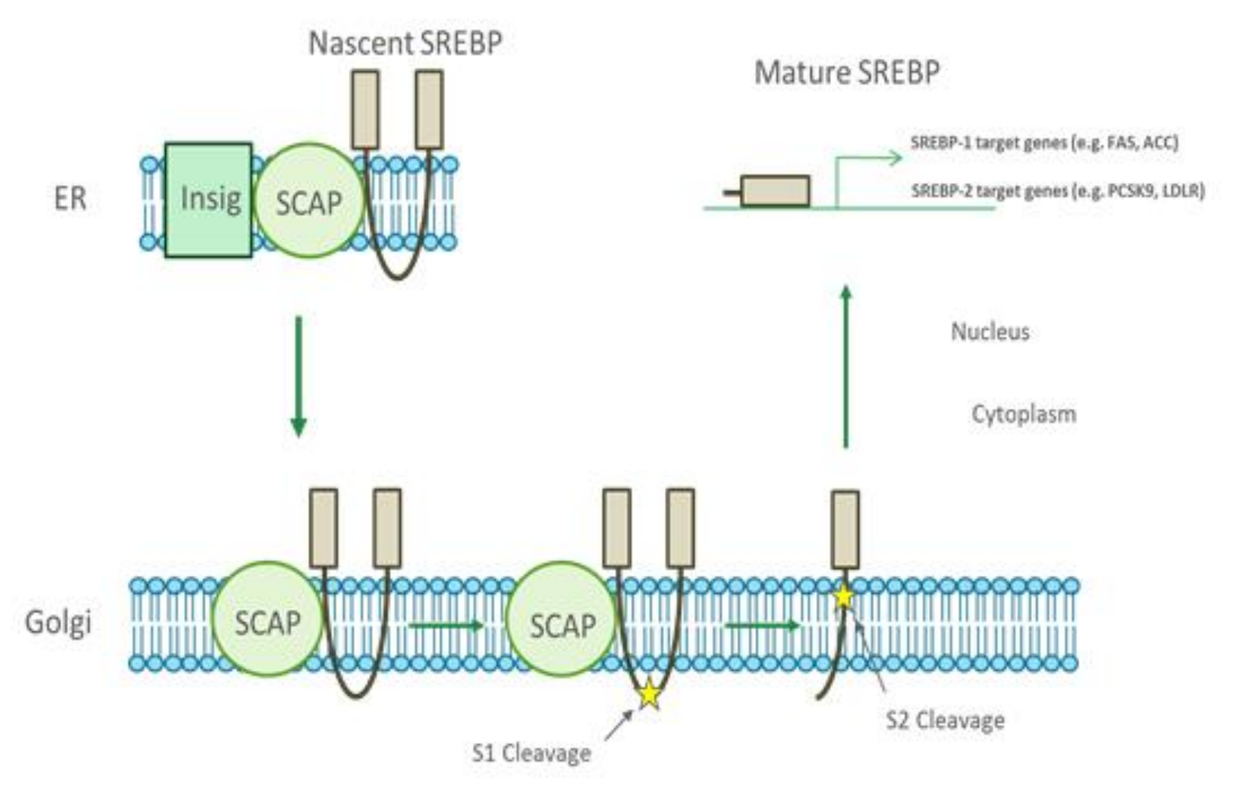
Nascent SREBP
Mature SREBP
ER
Insig
SCAP
Nucleus
Cytoplasm
Golgi
S1 Cleavage
S2 Cleavage

CAT-2003 and CAT-2054 are First in Class Inhibitors of SREBP, a Master Regulator of Lipid Metabolism

First in class SREBP inhibitors, acting on SREBP1 to inhibit key de novo lipogenesis genes, and acting on SREBP2 to inhibit key cholesterol synthesis genes

SREBP is upstream regulator of ACC and HMGCR

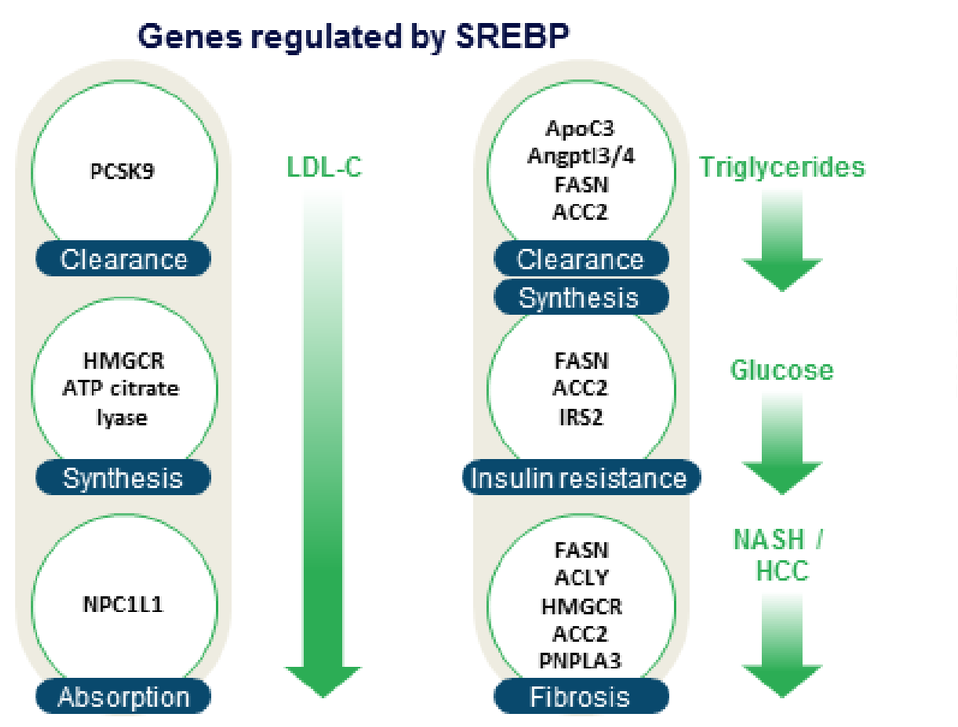
Genes regulated by SREBP
PCSK9
Clearance
HMGCR
ATP citrate lyase
Synthesis
NPC1L1
Absorption
LDL-C
ApoC3
Angptl3/4
FASN
ACC2
Clearance
Synthesis
FASN
ACC2
IRS2
Insulin resistance
FASN
ACLY
HMGCR
ACC2
PNPLA3
Fibrosis
Triglycerides
Glucose
NASH / HCC

CAT-2003 & CAT-2054 Inhibit ACC2 and HMGCR Expression

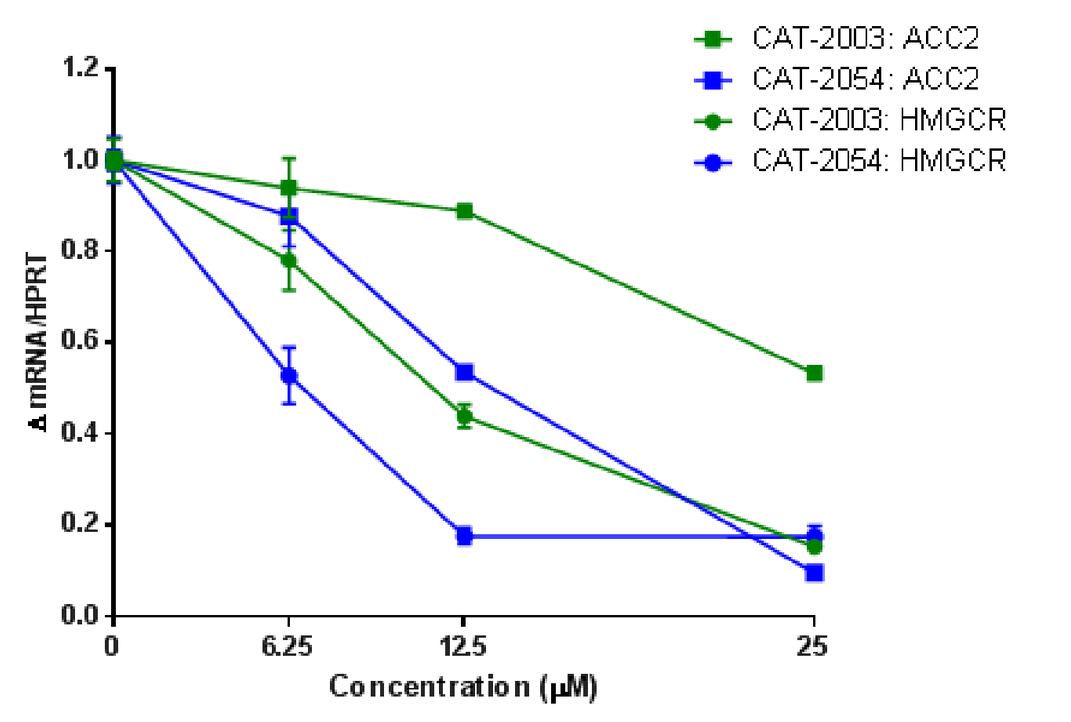
CAT-2003: ACC2
CAT-2054: ACC2
CAT-2003: HMGCR
CAT-2054: HMGCR
Δ mRNA/HPRT
Concentration (μM)

HepG2 cells were treated with the indicated concentrations of CAT-2003 or CAT-2054 and cells were harvested the next day for mRNA analysis. Total RNA was collected using RNeasy Plus Mini Kit (Qiagen #74136) and cDNA generated using SuperScript III (Invitrogen #18080-044) with random hexamers following the manufacturer's protocol. Relative mRNA expression levels were determined using TaqMan probes (Applied Biosystems, using recommended best primer pairs) with *HPRT* as internal control. Experiment was performed in triplicate. Error bars represent SEM.

Background
The Role of SREBP in NASH

- Non Alcoholic Fatty Liver Disease (NAFLD) is the consequence of accumulation of triglycerides in the liver. NAFLD is strongly associated with metabolic syndrome and patients diagnosed with NAFLD frequently also suffer from obesity, dyslipidemia, insulin resistance and hypertension.
- NASH is a more serious form of NAFLD which is characterized by inflammation in the liver which can further progress to include hepatocellular injury, chronic fibrosis, cirrhosis, hepatocellular carcinoma (HCC) and end-stage liver disease requiring transplantation.
- Sterol Response Element-Binding Proteins (SREBPs) are a family of basic helix-loop-helix nuclear transcription factors that regulate *de novo* synthesis of fatty acids and cholesterol in the liver.
- Insulin can regulate lipogenesis through SREBP-1c in the liver and hormone sensitive lipase in adipose tissue. Insulin activates SREBP-1c while insulin resistance is associated with defective inhibition of lipase activity. Combined, these activities result in the increased generation of free fatty acids (FFA) which are taken up by the liver and are associated with the development of hepatic steatosis and inflammation.
- The progression from NAFLD to NASH is highly variable and of unknown etiology although the presence of a mutation (I148M) in the patatin-like phospholipase domain containing 3 gene (PNPLA3) is associated with enhanced risk for progression from simple steatosis to steatohepatitis, cirrhosis and HCC. While it is not yet clear whether the mutation results in a gain or loss of endogenous function, the presence of an SREBP-1c-binding site in upstream promoter suggests that PNPLA3 is a SREBP-regulated gene.
- Accumulation of hepatic free cholesterol has been linked to hepatocyte injury and may be one of the pathological hits that drives progression to NASH. SREBP-2 regulates intracellular cholesterol biosynthesis and increased hepatic SREBP-2 has been observed in both rat and human NASH (Cabellero et all, 2009; Zhao, 2010).

Results and Summary

Study Design to Measure Efficacy of CAT-2003 Treatment in Murine Metabolic Model of NASH

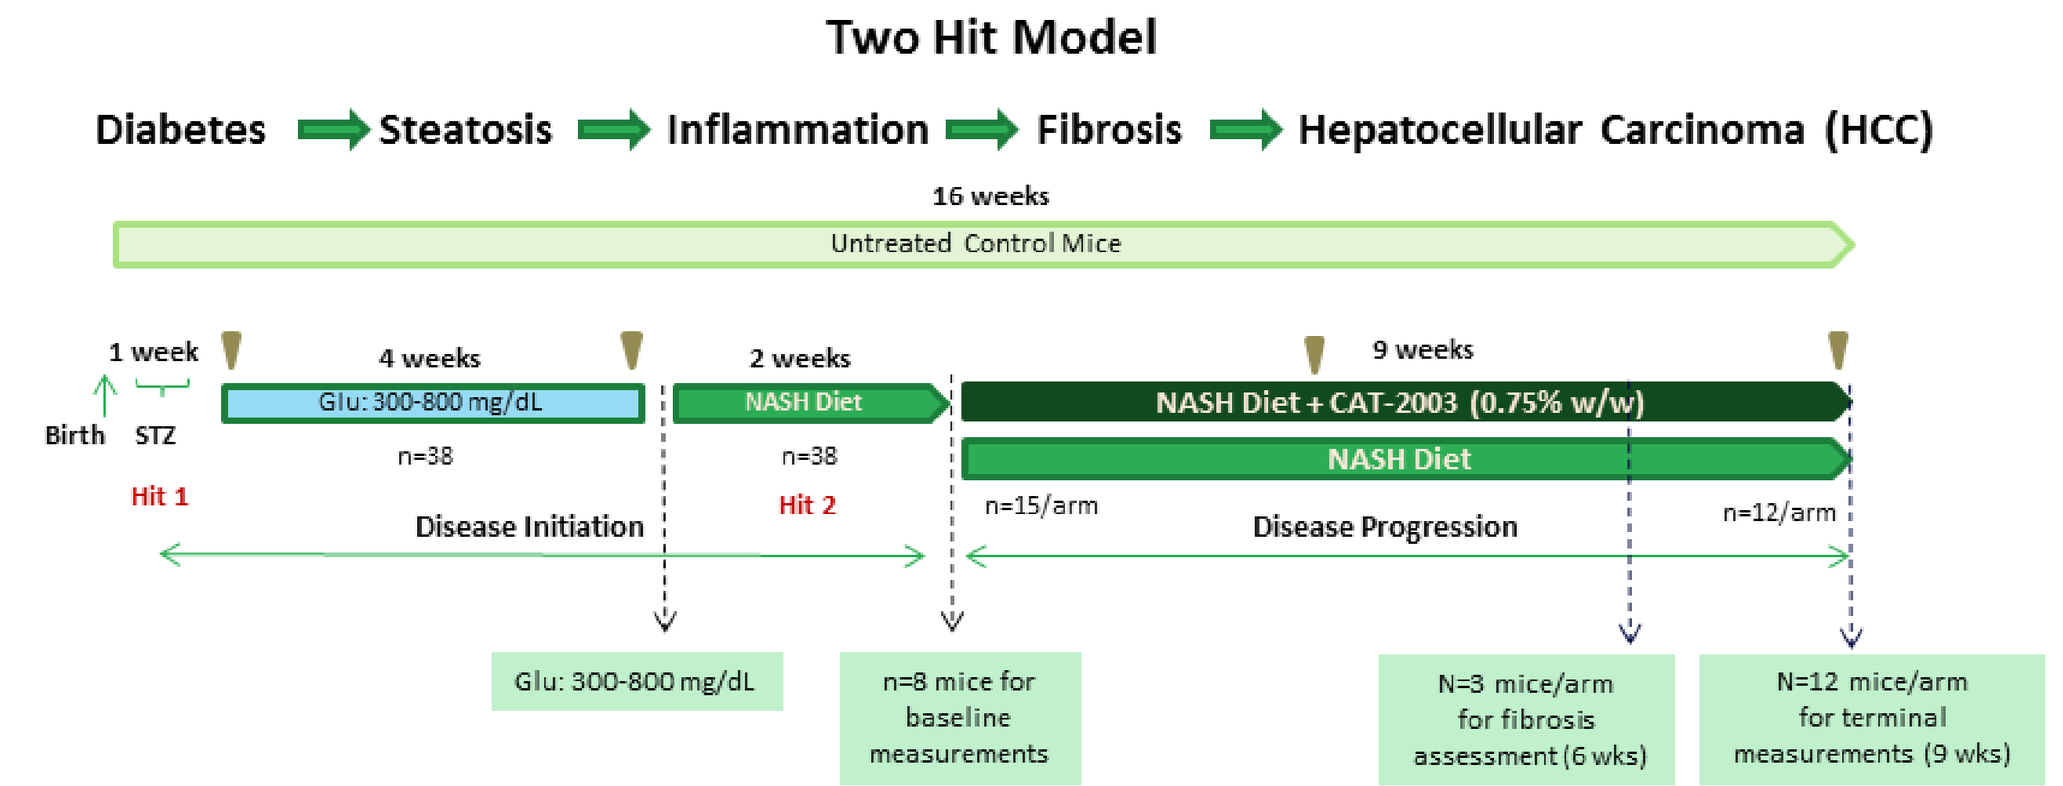
Two Hit Model
Diabetes → Steatosis → Inflammation → Fibrosis → Hepatocellular Carcinoma (HCC)
16 weeks
Untreated Control Mice
1 week
4 weeks
2 weeks
9 weeks
Birth
STZ
Glu: 300-800 mg/dL
NASH Diet
NASH Diet + CAT-2003 (0.75% w/w)
NASH Diet
n=38
n=38
n=15/arm
n=12/arm
Hit 1
Hit 2
Disease Initiation
Disease Progression
Glu: 300-800 mg/dL
n=8 mice for baseline measurements
N=3 mice/arm for fibrosis assessment (6 wks)
N=12 mice/arm for terminal measurements (9 wks)

C57BL/6NHsd timed pregnant female mice (E16) were obtained from Jackson Laboratories. Beginning at 2 days after birth, pups were administered Streptozotocin intraperitoneally at a dose of 200 mg/kg. 38 male mice with glucose levels between 300-800 mg/dL were enrolled in the study (disease cohort). A separate group of 8 male mice, maintained on the acclimation diet, was left untreated and assigned to the naïve control arm. At week 5, the mice in the disease cohort were switched to the modified high fat diet (mod-HFD) consisting of 40% kcal fat, 20% kcal fructose and 2% cholesterol for development of NASH. After 2 weeks on the NASH (mod-HFD) diet, 8 mice from the disease cohort were selected at random to define baseline levels for primary and secondary end-points in the study. The remainder of the disease cohort was randomized into either the disease control arm (n=15) or the CAT-2003 treatment arm (n=15). After 6 weeks of treatment (week 13), 3 satellite mice from the disease control arm and the CAT-2003 arm were sacrificed to determine the extent of disease progression. The final study groups including the naïve controls (n=8) and the two disease arms (n=12 each) were terminated at week 16 (after 9 weeks of treatment with CAT-2003).

CAT-2003 Treatment Significantly Abrogates the NASH Diet Induced Increase in Liver Weight without Significantly Affecting Body Weight

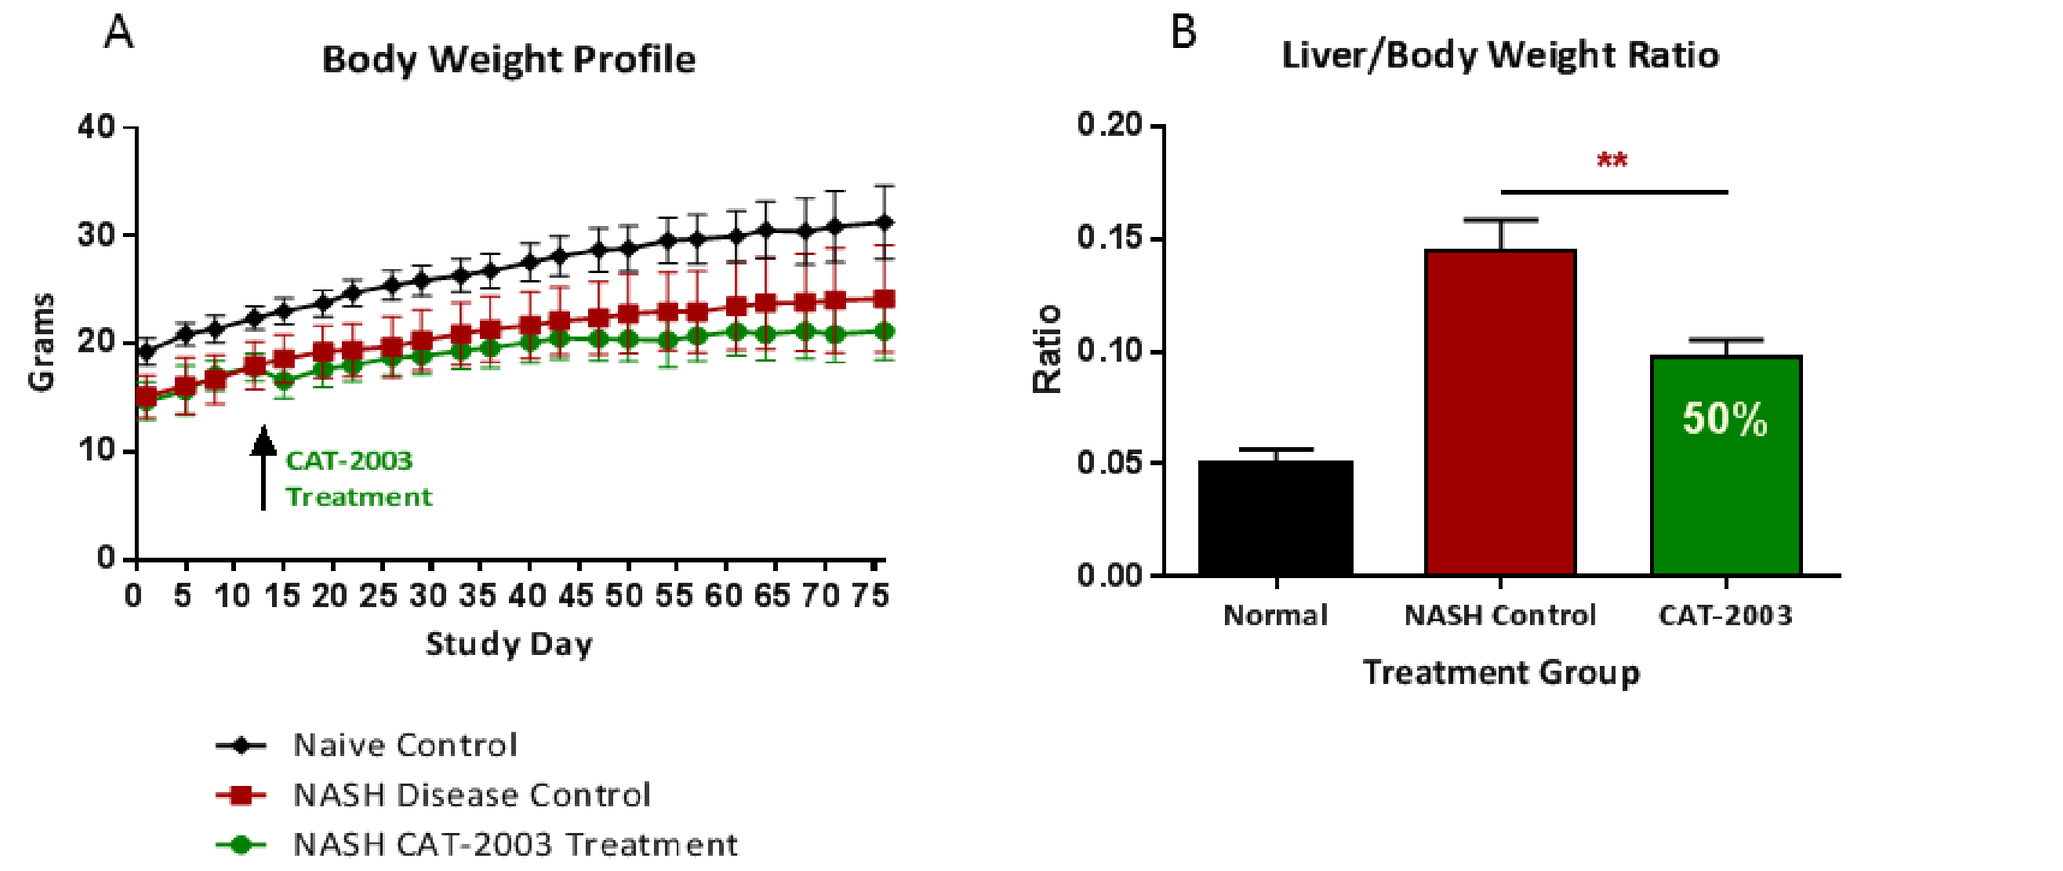
A
Body Weight Profile
Grams
Study Day
CAT-2003 Treatment
Naive Control
NASH Disease Control
NASH CAT-2003 Treatment
B
Liver/Body Weight Ratio
Ratio
**
50%
Normal
NASH Control
CAT-2003
Treatment Group

Panel A: body weights of mice in the naïve control group (n=8) and in the NASH disease control group (n=12) and NASH CAT-2003 treatment group (n=12). Data is represented as mean body weight at each measurement±SEM. **Panel B**: the data represents the mean±SEM of the ratio of liver weight to body weight (g/g) for each mouse at termination (week 16). **p<0.01 ANOVA with Tukey post test for multiple comparisons.

CAT-2003 Treatment Significantly Reduces Hepatic Triglyceride and Cholesterol Levels at 16 Weeks

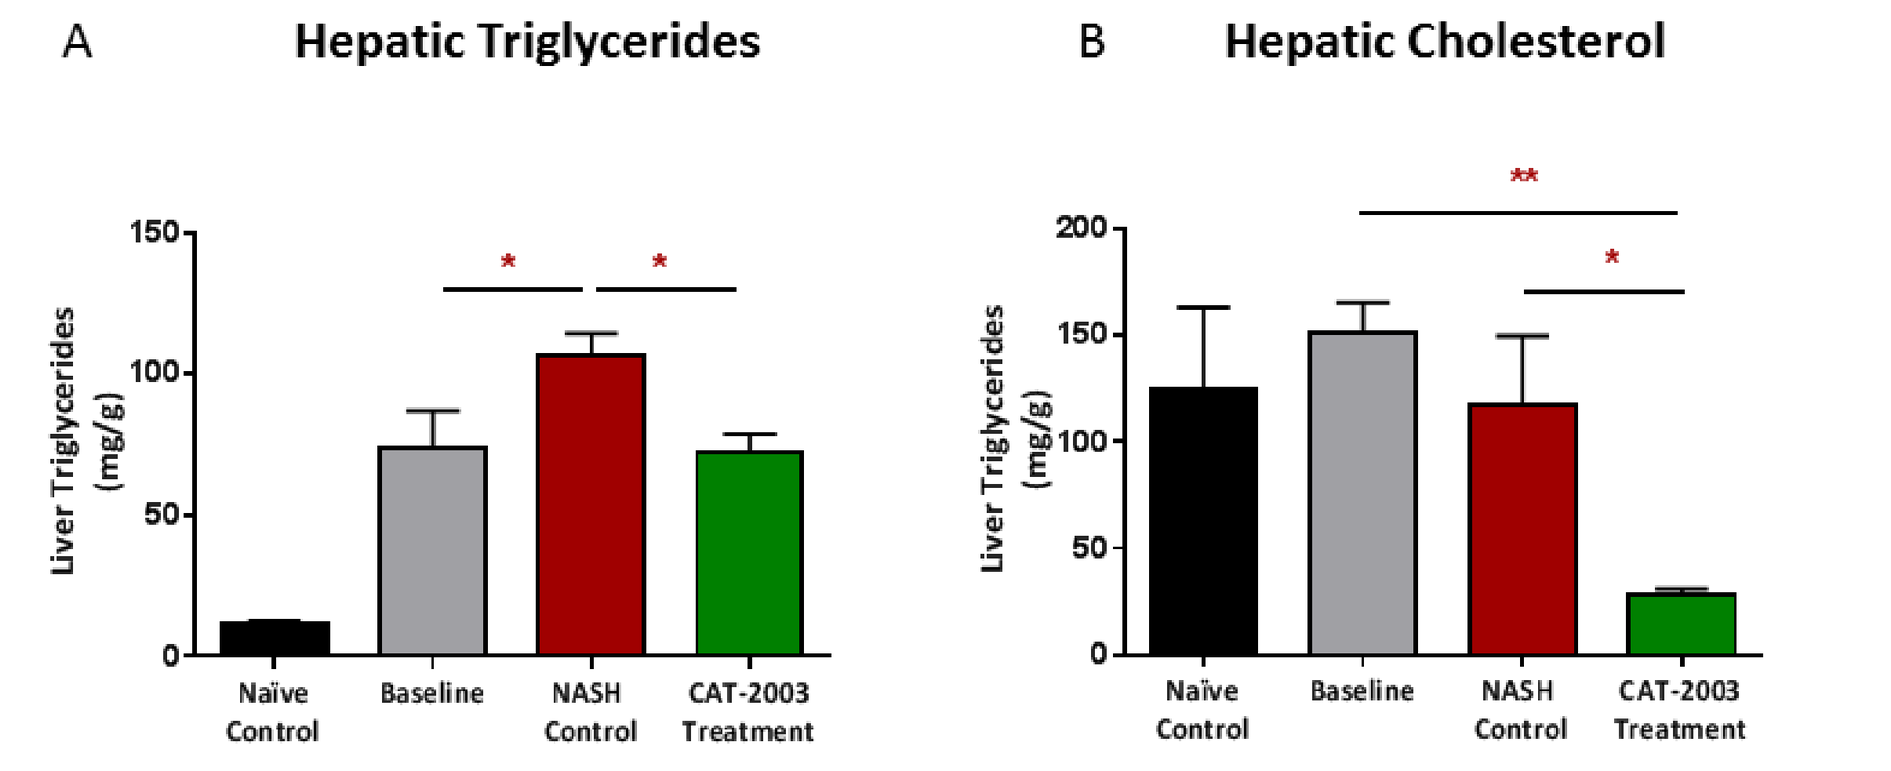
A
Hepatic Triglycerides
Liver Triglycerides (mg/g)
B
Hepatic Cholesterol
Naïve Control
Baseline
NASH Control
CAT-2003 Treatment

Liver total cholesterol (free cholesterol + cholesterol esters) and triglyceride levels were determined from frozen samples obtained at termination using assay kits from Biovision (Milipitas, CA) per manufacturer's instructions. The data is expressed as the mean mg triglyceride **Panel A** or mean mg cholesterol **Panel B** per gram of total protein in the liver sample±SEM. Baseline (n=8) samples were collected at week 7, naïve (n=8), disease control (n=12) and CAT-2003 treatment (n=12) samples were collected at week 16. *p<0.05, **p<0.01; ANOVA with Tukey post test for multiple comparisons. Plasma cholesterol levels were significantly increased in the NASH control mice (165%) and CAT-2003 treated mice (104%) relative to the naïve control mice at week 16 (data not shown).

CAT-2003 Treatment Results in Reduced Hepatic Steatosis, Inflammation and Injury (NAS)

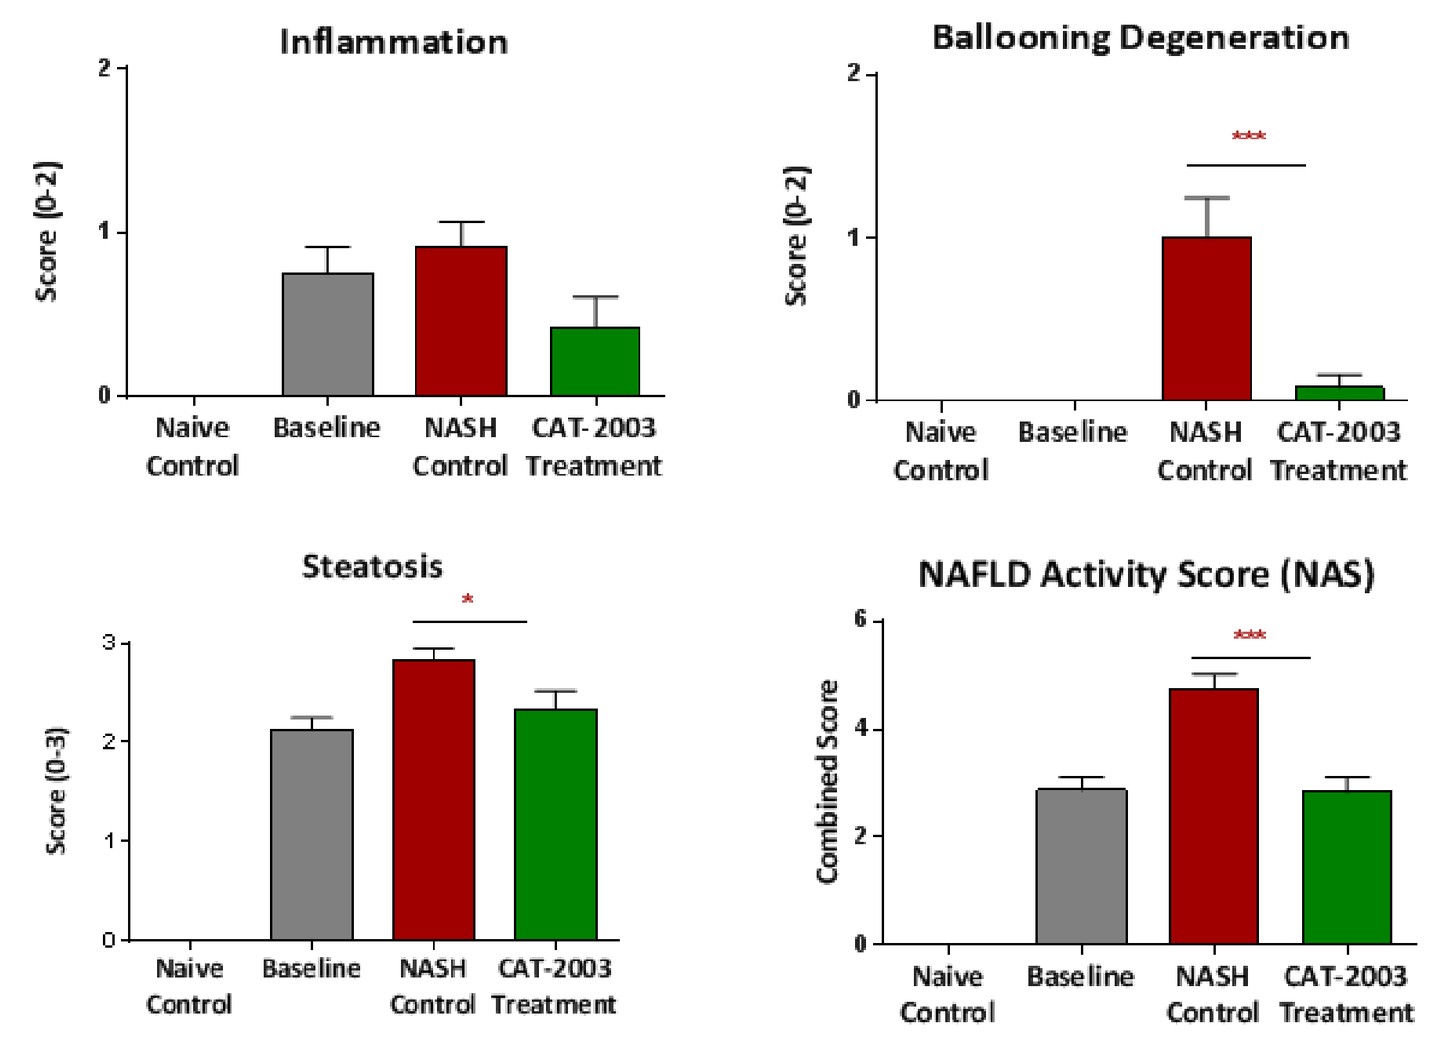
Inflammation
Ballooning Degeneration
Steatosis
NAFLD Activity Score (NAS)
Score (0-2)
Score (0-3)
Combined Score
Naive Control
Baseline
NASH Control
CAT-2003 Treatment

FFPE tissues were cut into 6mm sections and stained using hematoxylin and eosin. Stained slides were scored in blinded fashion using the guidelines of the American Association for Study of Liver Disease.* The NASH Activity Score is the sum of the individual score for steatosis, inflammation and degeneration for each animal. Data is expressed as the mean score±SEM. *p<0.05, ***p<0.001; ANOVA with Tukey post test for multiple comparisons. **Brunt EM, Janney CG, DiBisceglie AM, Neuschwander-Tetri BA, Bacon BR. Nonalcoholic steatohepatitis: a proposal for grading and staging the histological lesions. Am. J. Gastroenterol. 1999; 94:2467-2474*

Treatment with CAT-2003 Reduced Steatohepatitis and Ballooning Degeneration

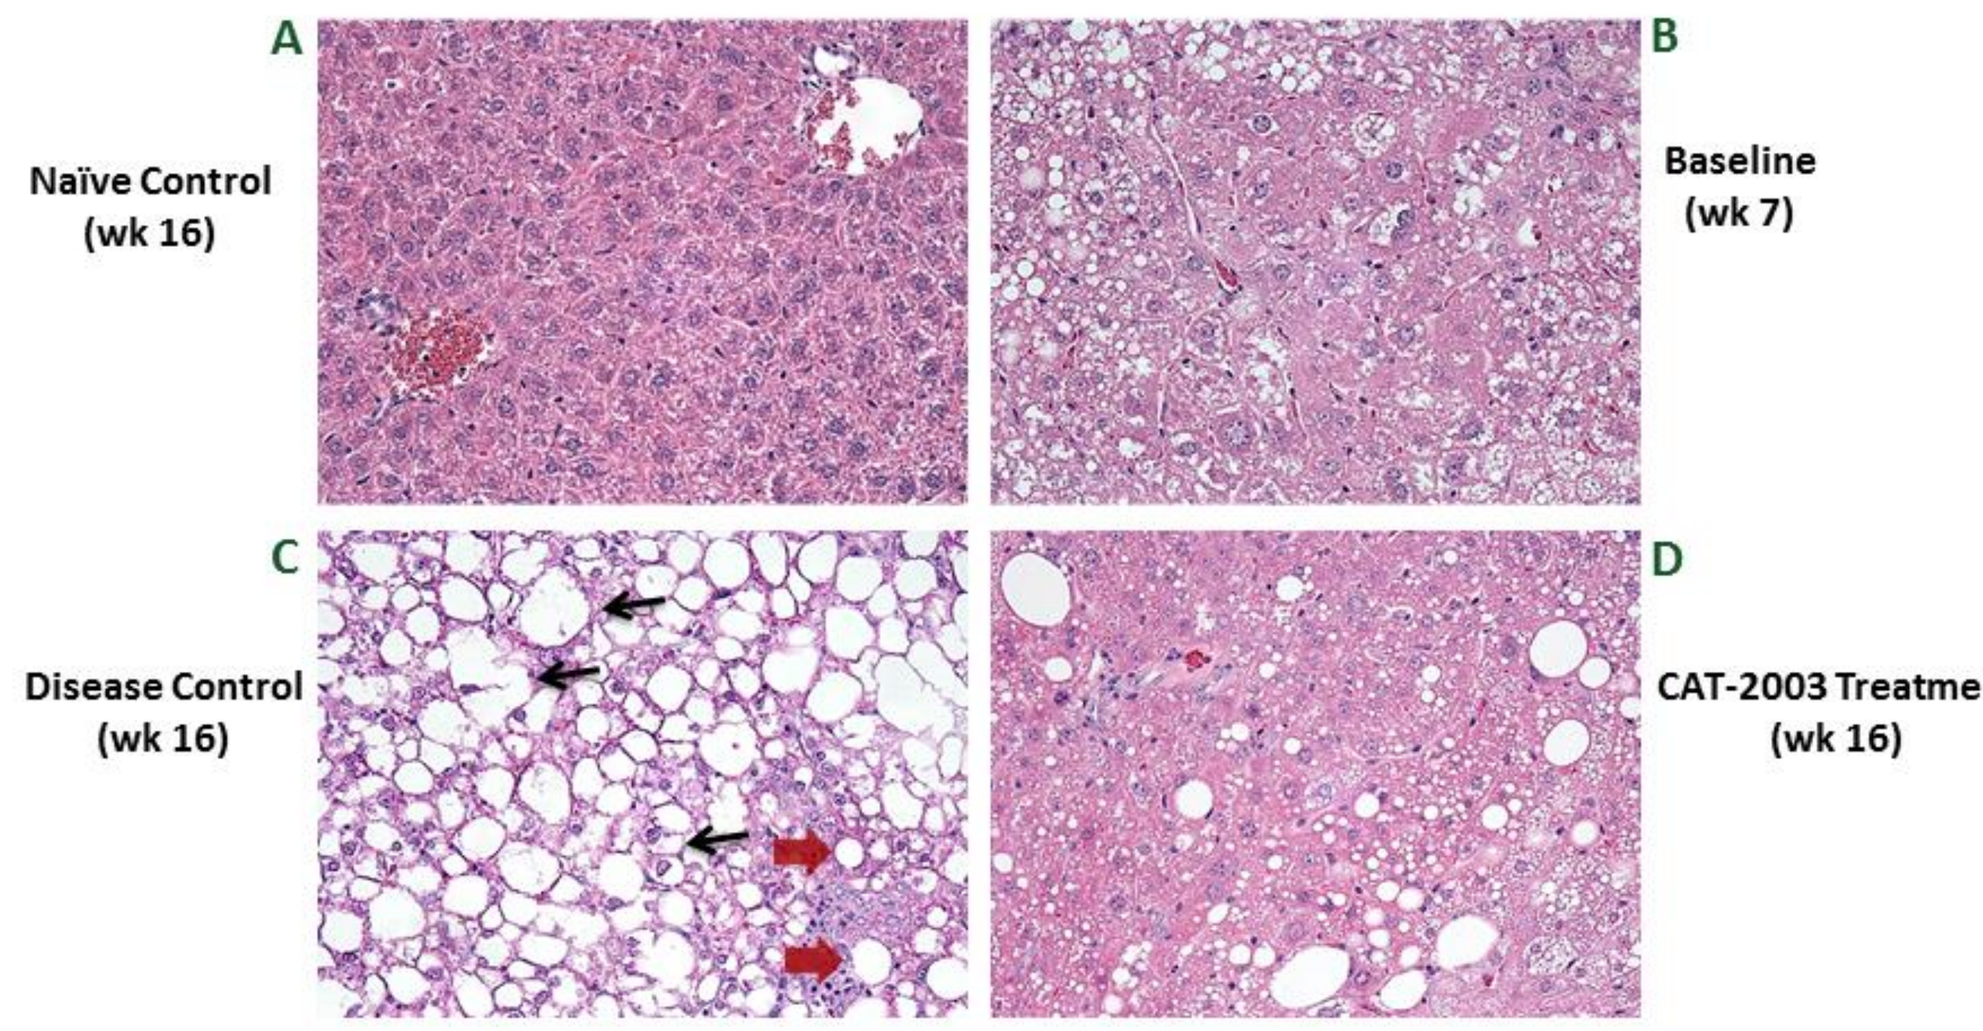
H&E Sections from Formalin-Fixed Paraffin-Embedded Liver Tissue
A
Naïve Control (wk 16)
B
Baseline (wk 7)
C
Disease Control (wk 16)
D
CAT-2003 Treatment (wk 16)

Representative H&E stained sections of liver (40X magnification). **Panel A**: section from liver of naïve control animal collected at week 16; **Panel B**: section from baseline animal collected at week 7 (2 weeks on NASH diet); **Panel C**: section from disease control animal at week 16, red arrows () indicate steatotic hepatocytes, black arrows () indicate ballooning degeneration; **Panel D**: section from disease group treated with CAT-2003 collected at week 16.

Treatment with CAT-2003 Results in Significantly Reduced Liver Fibrosis relative to NASH Control

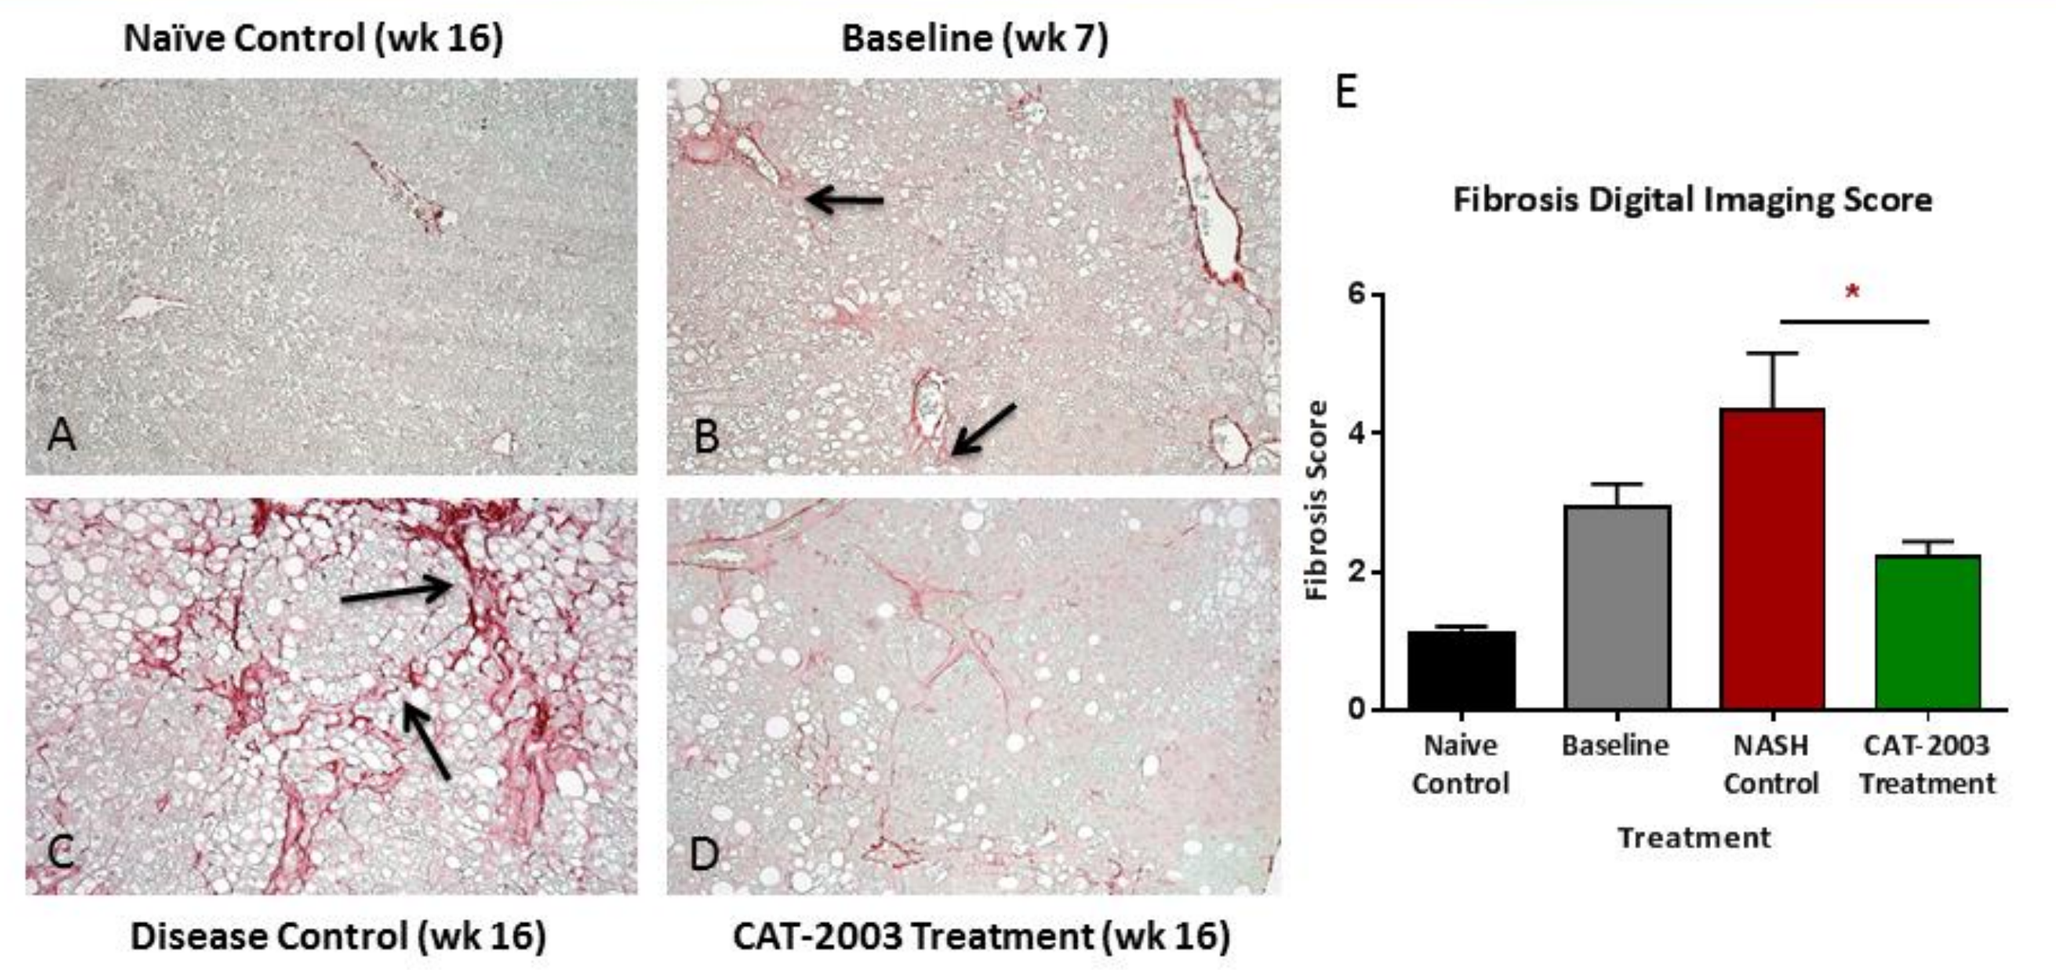
Naïve Control (wk 16)
Baseline (wk 7)
Disease Control (wk 16)
CAT-2003 Treatment (wk 16)
E
Fibrosis Digital Imaging Score
Fibrosis Score
Naive Control
Baseline
NASH Control
CAT-2003 Treatment
Treatment

FFPE liver was cut into 6 μm sections and stained with picrosirius red to visualize collagen deposition (20X magnification). The extent of collagen staining was assessed by two independent methods: scoring in blinded fashion by a pathologist (data not shown) and digital quantitation of the signal (Reveal Biosciences, San Diego CA). **Panel A**: section from liver of naïve control animal collected at week 16; **Panel B**: section from baseline animal collected at week 7, arrows indicate perivascular fibrosis; **Panel C**: section from disease control animal at week 16, arrows indicate bridging fibrosis; **Panel D**: section from disease group treated with CAT-2003 collected at week 16. *p<0.05, ANOVA with Tukey post test for multiple comparisons. **Panel E**: Digital score data expressed as the mean±SEM; n=8 (naïve control & baseline), n=12 (NASH control & CAT-2003 treatment).

CAT-2003 Modulates Multiple Pathways of Metabolism, Inflammation and Fibrosis Which Play Important Roles in the Pathogenesis of NASH

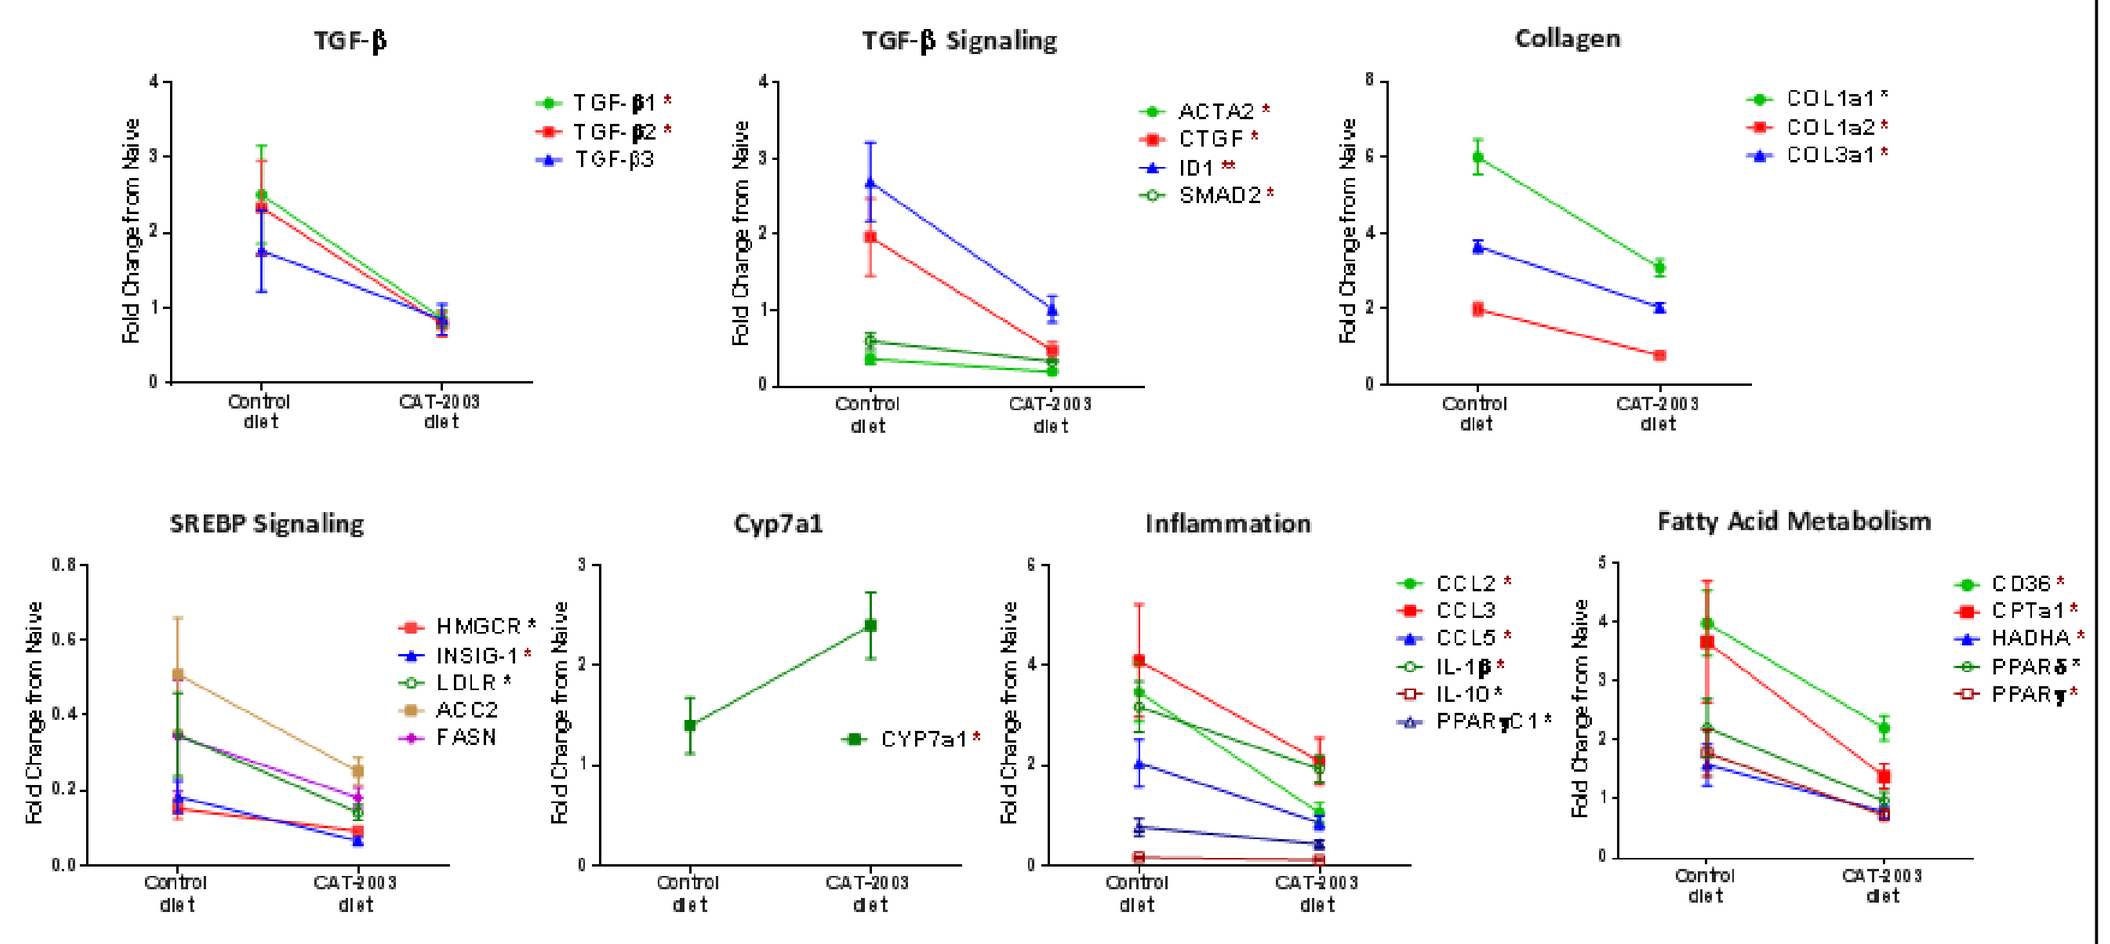
TGF-β
TGF-β Signaling
Collagen
SREBP Signaling
Cyp7a1
Inflammation
Fatty Acid Metabolism
Fold Change from Naive
Control diet
CAT-2003 diet

RNA was extracted from approximately 10 mg pieces of frozen liver tissue and homogenized in Qiagen RLT buffer using a tissue microhomogenizer. The homogenized sample was clarified by centrifugation and passed through a Qiagen gDNA elimination column. cDNA was generated using SuperScript III (Invitrogen #18080-044) with random hexamers following the manufacturer's protocol. Relative mRNA expression levels were determined using TaqMan probes (Applied Biosystems, using recommended best primer pairs) with *HPRT* as internal control. Data is expressed as the mean fold change in RNA expression relative to naïve (n=12 for both groups). Error bars represent SEM. **p<0.01, *p<0.05, two-tail t test; *p<0.05 one-tail t test.

CAT-2003 Treatment Significantly Reduces Development of Pre-neoplastic Lesions in the Liver

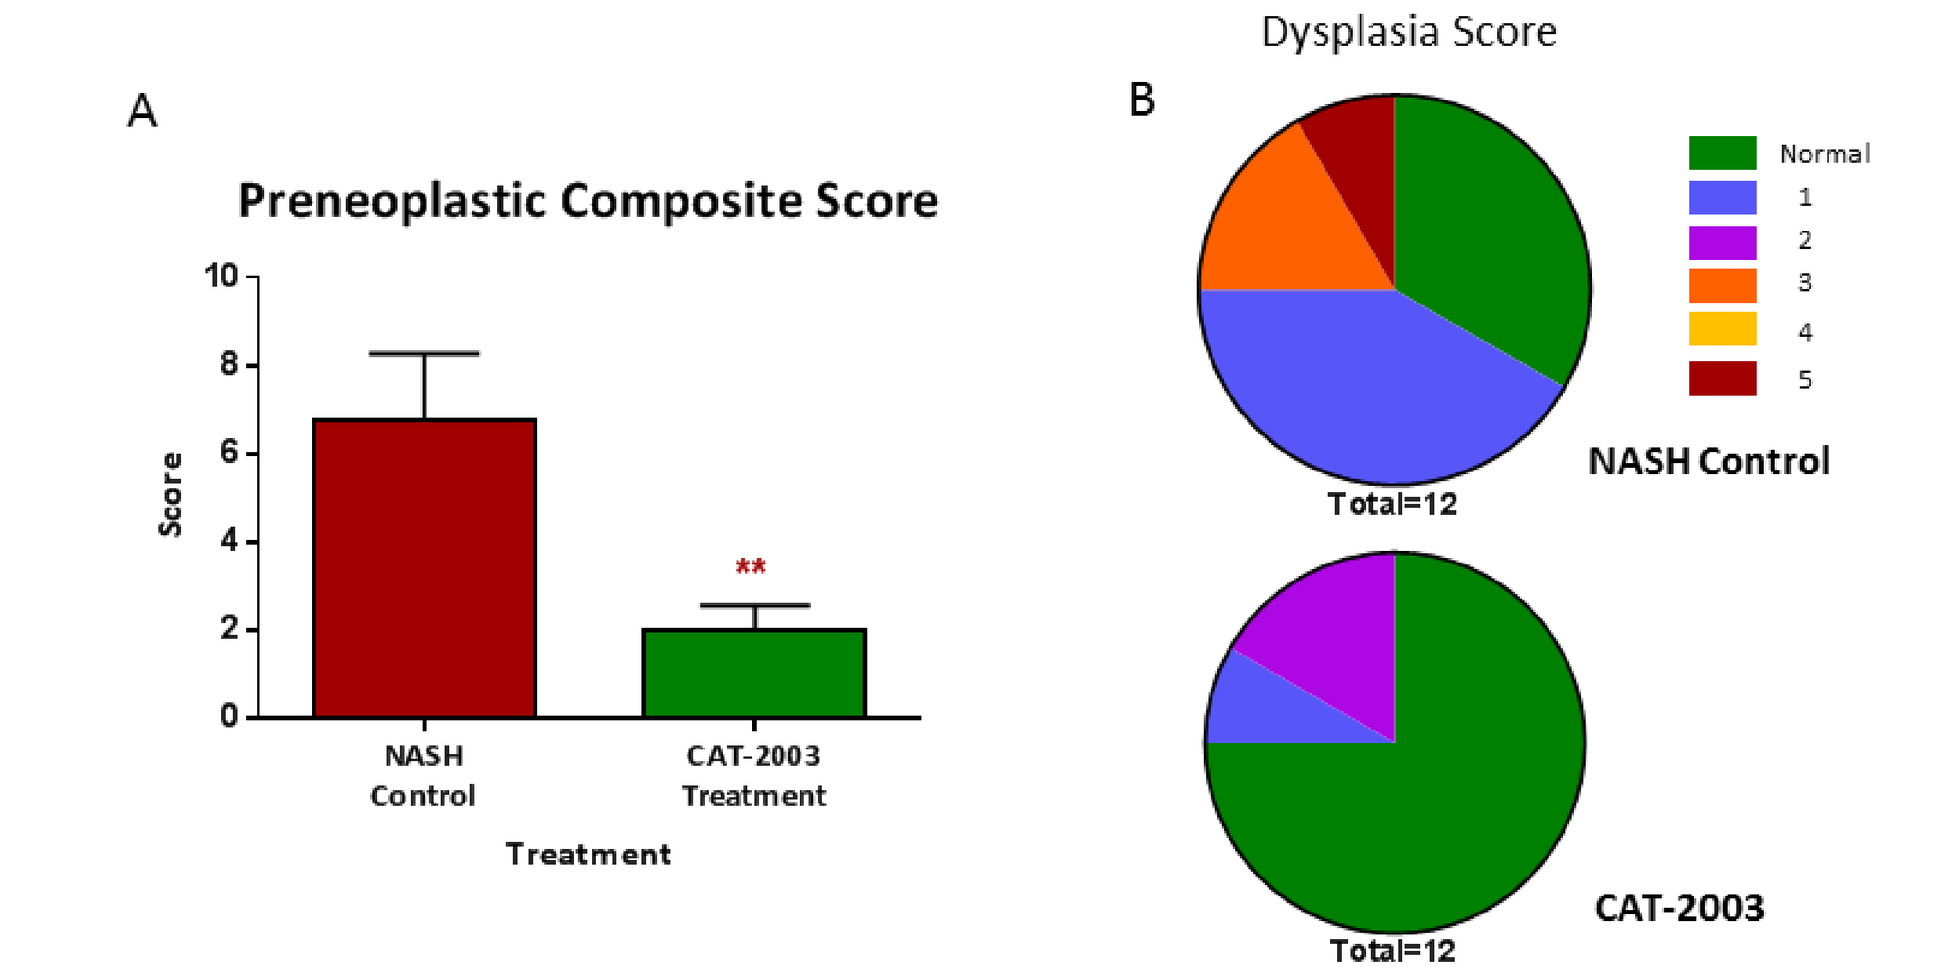
A
Preneoplastic Composite Score
Score
NASH Control
CAT-2003 Treatment
Treatment
B
Dysplasia Score
Normal
NASH Control
Total=12
CAT-2003
Total=12

H&E sections from FFPE liver were evaluated and scored in blinded fashion by a board certified pathologist. **Panel A**: The pre-neoplastic composite score is a sum of separate pre-neoplastic lesion scores: foci of cellular alteration, clear cell focus of cellular alteration, dysplastic focus (cluster of atypical hepatocytes without discrete margin; usually smaller than a lobule) and dysplastic nodule (atypical hepatocytes larger than a lobule and with discrete border). The data is expressed as the mean score±SEM. **p<0.01 two-tail t test. **Panel B**: The dysplasia score is the combination of the dysplastic focus score and dysplasia nodule score. The data is expressed as the fraction of each severity score.

Summary and Conclusions

- In this murine model of NASH, all disease control mice developed characteristic features of NASH including hepatic steatosis, lobular inflammation, ballooning degeneration, and fibrosis.
- CAT-2003 treatment resulted in decreased expression of genes involved in fibrosis (TGFβ production, TGFβ signaling and collagen) and inflammation (CCL2, CCL5, IL-1β). In addition CAT-2003 mediated inhibition of SREBP resulted in decreased expression of HMGCR, the rate limiting step in cholesterol synthesis, and increased expression of CYP7a1, the rate limiting step in bile acid synthesis potentially resulting in increased clearance of cholesterol from the liver by conversion to bile acids.
- The SREBP-mediated reduction in hepatic cholesterol combined with reduced expression of NF-kB mediated pro-inflammatory gene expression may contribute to the efficacy observed in this study by suppressing the activation of hepatic stellate cells and the recruitment of activated macrophages to the liver.
- CAT-2003 treatment demonstrated therapeutic activity across multiple pathologic pathways to achieve significant reductions in disease severity including the NAFLD Activity Score (NAS), hepatic cholesterol levels, fibrosis and overall severity of early hepatocellular carcinoma.
- CAT-2003 reduced the multiplicity (type and number) of pre-neoplastic lesions (foci of cellular alteration, dysplastic foci and nodules), suggesting a potential benefit for delaying or preventing NASH-associated HCC.
- Phase 1 studies with both CAT-2003 and CAT-2054 demonstrated that systemic exposures above the efficacious level in this mouse study could be achieved with doses which were safe and well tolerated (Donovan, J., et al. 2016. National Lipid Association. Abstract 181: Phase 1 Multiple Ascending Dose Study of CAT-2054, a Novel Oral Sterol Regulatory Element Binding Protein Inhibitor).
- CAT-2054 was designed to be more resistant than CAT-2003 to hydrolysis in the intestine resulting in approximately 10-fold more delivery of intact CAT-2054 to the liver relative to CAT-2003 for a given dose (Vu, C. B. et al. Synthesis and Characterization of Fatty Acid Conjugates of Niacin and Salicylic Acid. *J. Med. Chem.* **2016**, *59*, 1217-1231).
- CAT-2054 is currently in a phase 2a study with LDL-C lowering as the primary end-point.
- The efficacy observed with CAT-2003 strongly supports future clinical investigation of CAT-2054, chosen for its superior PK profile and liver exposure levels, for treatment of NASH.

catabasis
a return to health